Poster M262

Effects of Efgartigimod Treatment on Humoral and Cellular Immune Responses: Analysis of T-Cell–Dependent Antibody Response in Cynomolgus Monkeys

Jenna Casey,[1] Mario Cocco,[2] Daniel Thwaites,[2] Christopher Cooper,[2] Gordon McPhie,[2] Ornella Binazon,[1] Mahan Moshir,[1] Peter Vanhoenacker,[1] Dieter Defever,[1] Sofie Priem,[1] Christa Kneip,[1] Sophie Steeland,[1] Gwenda Pynaert,[1] Peter Ulrichts,[1] Judith Baumeister[1]

[1]argenx, Ghent, Belgium; [2]Labcorp Early Development Laboratories Ltd., Harrogate, UK

SCAN ME

INTRODUCTION

Efgartigimod Mechanism of Action: Blocking FcRn

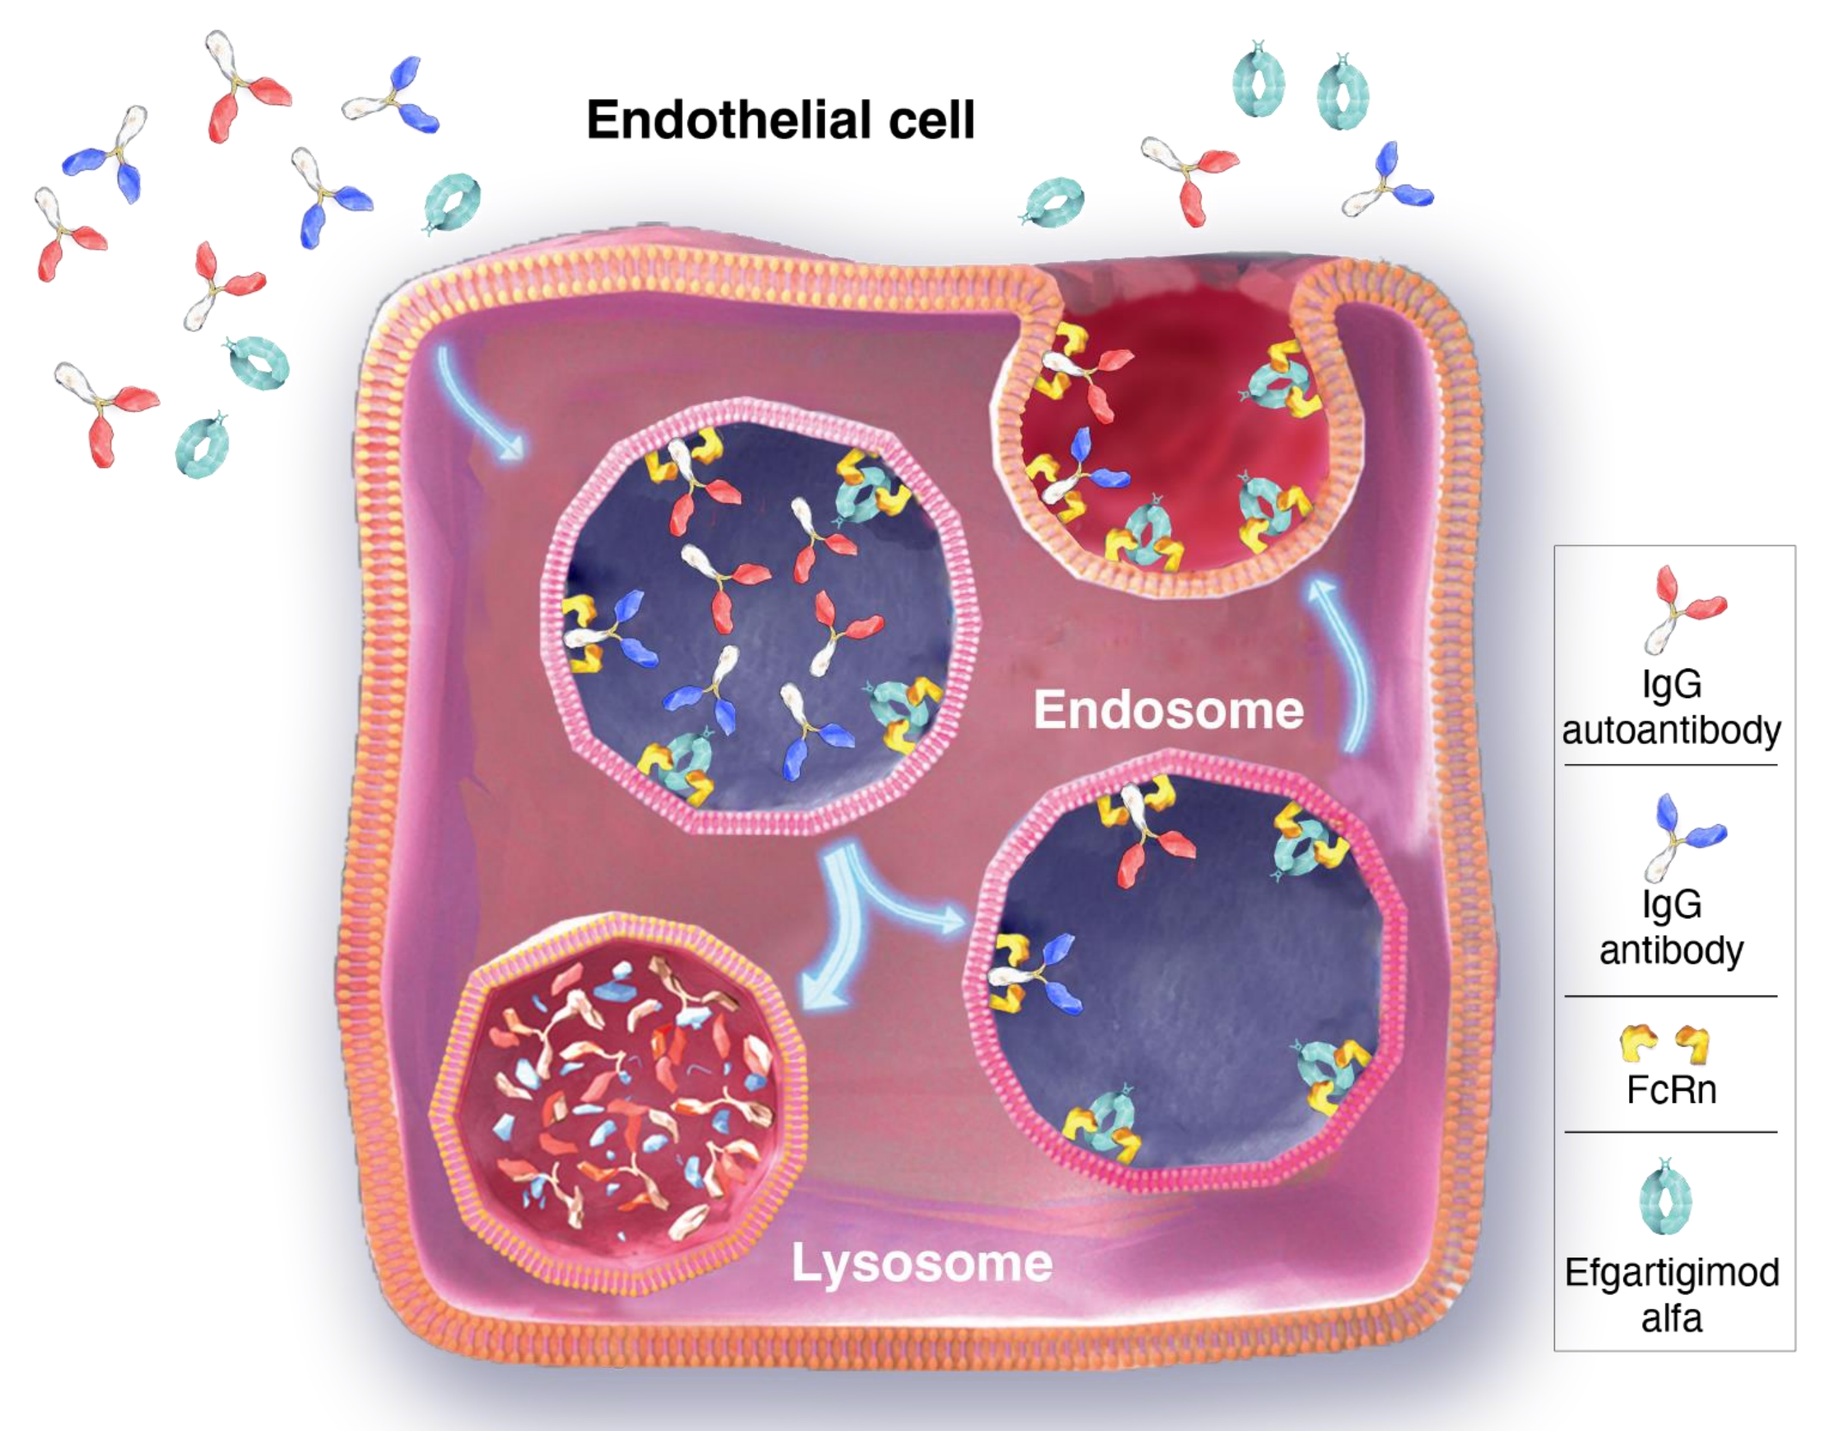

Endothelial cell
Endosome
Lysosome
IgG autoantibody
IgG antibody
FcRn
Efgartigimod alfa

- FcRn recycles IgG, extending its half-life and maintaining its serum concentration[1]
- Efgartigimod is a human IgG1 Fc fragment, a natural ligand of FcRn, engineered for increased affinity to FcRn[2,3]
- Efgartigimod was designed to outcompete endogenous IgG, preventing recycling and promoting IgG lysosomal degradation without directly impacting its production[2-6]
 - Targeted reduction of all IgG subtypes
 - No impact on IgM or IgA
 - No reduction in albumin levels
 - No increase in cholesterol

Immunosuppressive Therapies Used in Treatment of gMG May Increase Susceptibility to Infections and Impair Immune Response to Vaccines[7-8]

- Previous gMG treatments have the potential to[7,9-10]:
 - Increase risk of infection
 - Reduce therapeutic antibody response
 - Impair cellular immune responses to vaccination
- Glucocorticoids and B-cell–depleting therapies (including MMF) have been shown to substantially reduce immunogenicity of mRNA vaccines to SARS-CoV-2[8]

Evidence Suggests Efgartigimod Does Not Impair Humoral Response to COVID-19 Vaccination[11]

- Among ADAPT+ study patients with gMG who received COVID-19 vaccination prior to October 2021, COVID-19 immunization resulted in antigen-specific IgG responses in most patients receiving efgartigimod (12/15 patients)[11,12]
- Impact of concomitant immunosuppressive therapies (eg, glucocorticoids and B-cell–depleting therapies such as MMF) warrants additional investigations[11]

Keyhole Limpet Hemocyanin (KLH): A Model Antigen for Testing T-Cell–Dependent Antibody Response (TDAR) and Simulating Vaccines Responses[13]

- Following KLH immunization, PBMCs secrete cytokines including IFN-γ as part of cell-mediated immune response[13]
- TDAR against KLH has been demonstrated in cynomolgus monkeys[14]
- Quantification of the antibody response to immunization with a T-cell–dependent antigen is a sensitive method for assessing immunocompetence[14]

METHODS

Outcomes Reported Include Timing and Magnitude of Anti-KLH IgM and IgG Responses and Elicited Levels of IFN-γ Secretion of PBMCs in Response to KLH Challenges in an ELISpot Assay

Study Design: TDAR in Cynomolgus Monkeys

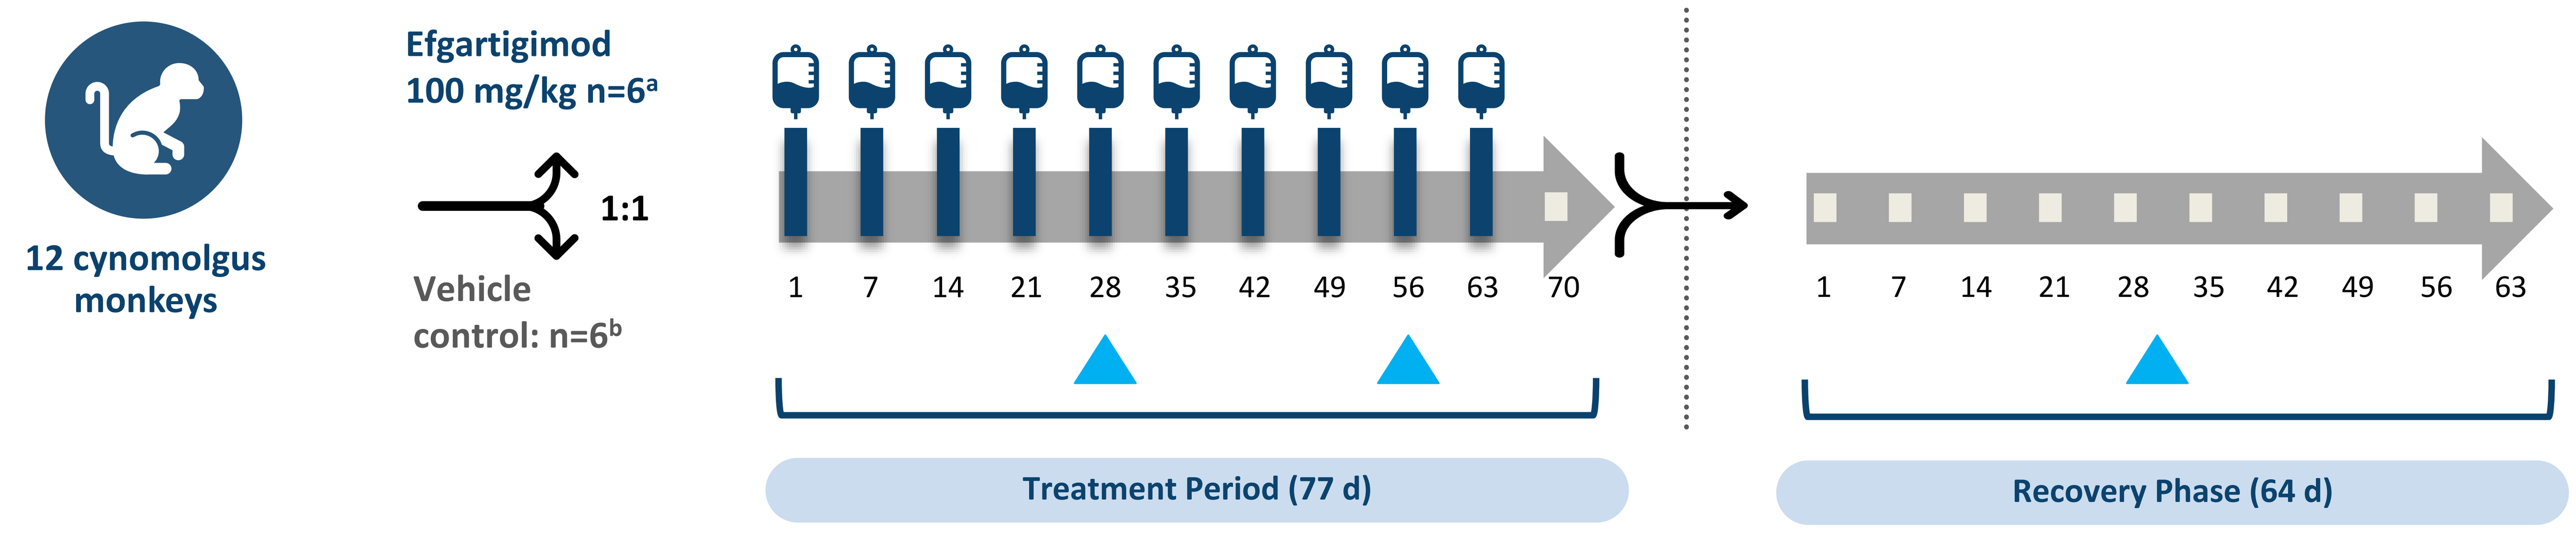

12 cynomolgus monkeys
Efgartigimod 100 mg/kg n=6[a]
1:1
Vehicle control: n=6[b]
1 7 14 21 28 35 42 49 56 63 70
1 7 14 21 28 35 42 49 56 63
Treatment Period (77 d)
Recovery Phase (64 d)

Efgartigimod/vehicle control intravenous administration

KLH (10 mg) subcutaneous administration

[a]The study group includes both male (n=3) and female (n=3) cynomolgus monkeys. [b]The vehicle control article was 20 mM histidine/histidine HCl, 60 mM sucrose, 100 mM sodium chloride, 10 mM L-methionine 0.04% w/v polysorbate 80, pH 6.0.

SUMMARY

- Lower anti-KLH IgG titers were observed after the second KLH challenge in efgartigimod-treated animals that normalized during the treatment-free period
- No difference was observed in IgM titers or KLH-elicited cellular response between efgartigimod- and vehicle control–treated animals
- T-cell–dependent antibody and cellular immune responses were mounted to a prototypical antigen under efgartigimod treatment
- Total IgG titers significantly decreased under efgartigimod treatment, consistent with the mechanism of action of the drug

RESULTS

Humoral Response: Anti-KLH IgM and IgG Antibodies Titers

Figure 1. Anti-KLH IgG Titer – Average Male + Female

- Anti-KLH IgG: lower IgG titers after 2nd KLH dose in efgartigimod-treated animals; however, no statistical difference between groups after washout

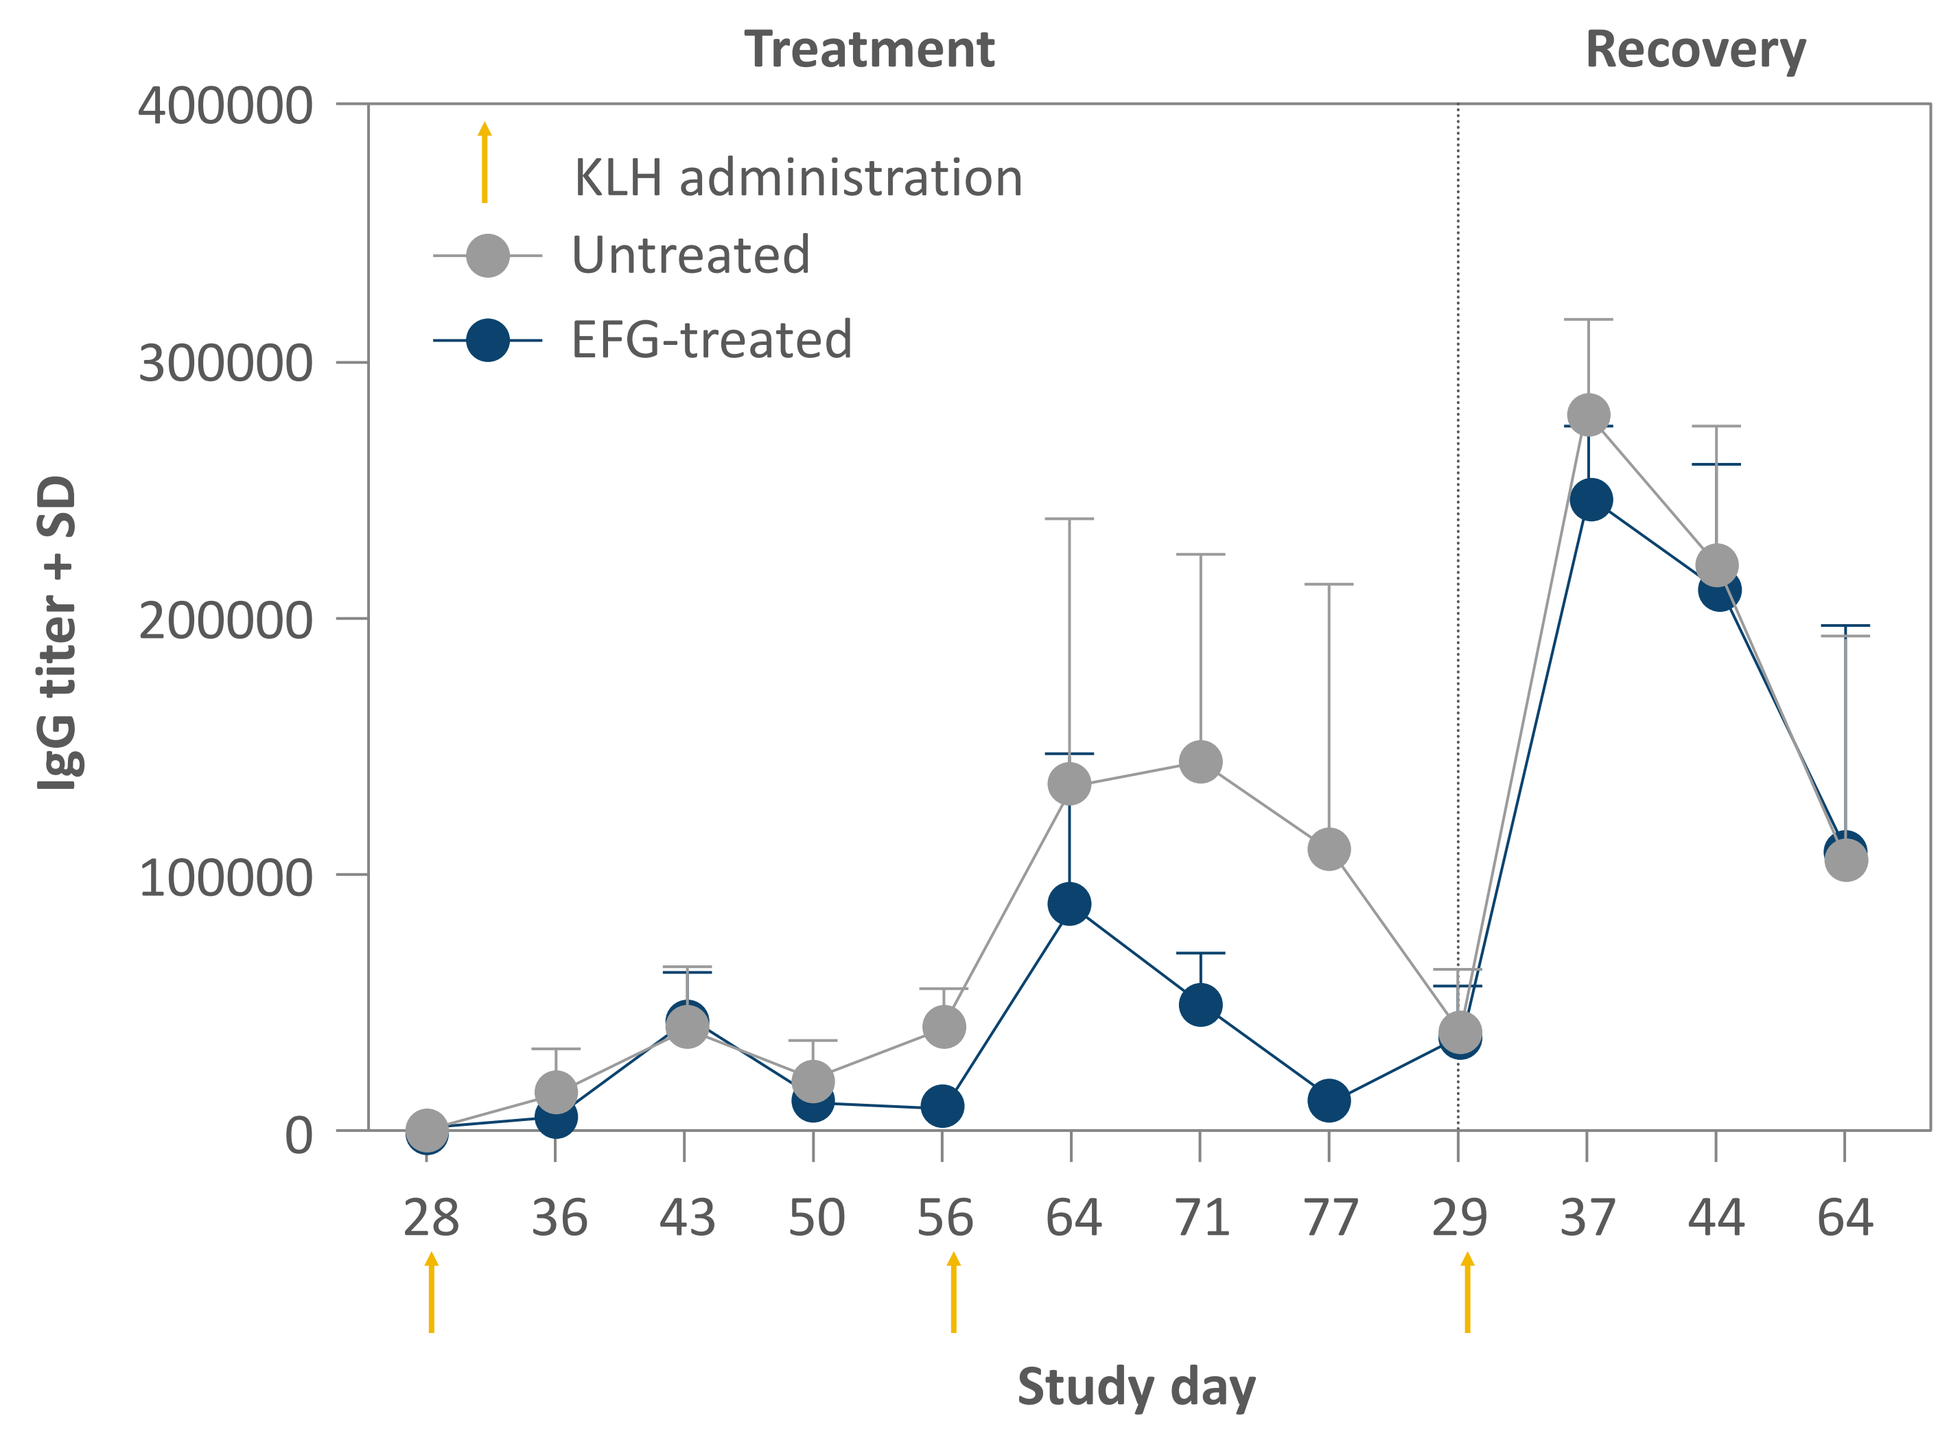

Treatment
Recovery
KLH administration
Untreated
EFG-treated
IgG titer + SD
400000
300000
200000
100000
0
28 36 43 50 56 64 71 77 29 37 44 64
Study day

Figure 2. Anti-KLH IgM Titer – Average Male + Female

- Anti-KLH IgM: no statistical differences between groups

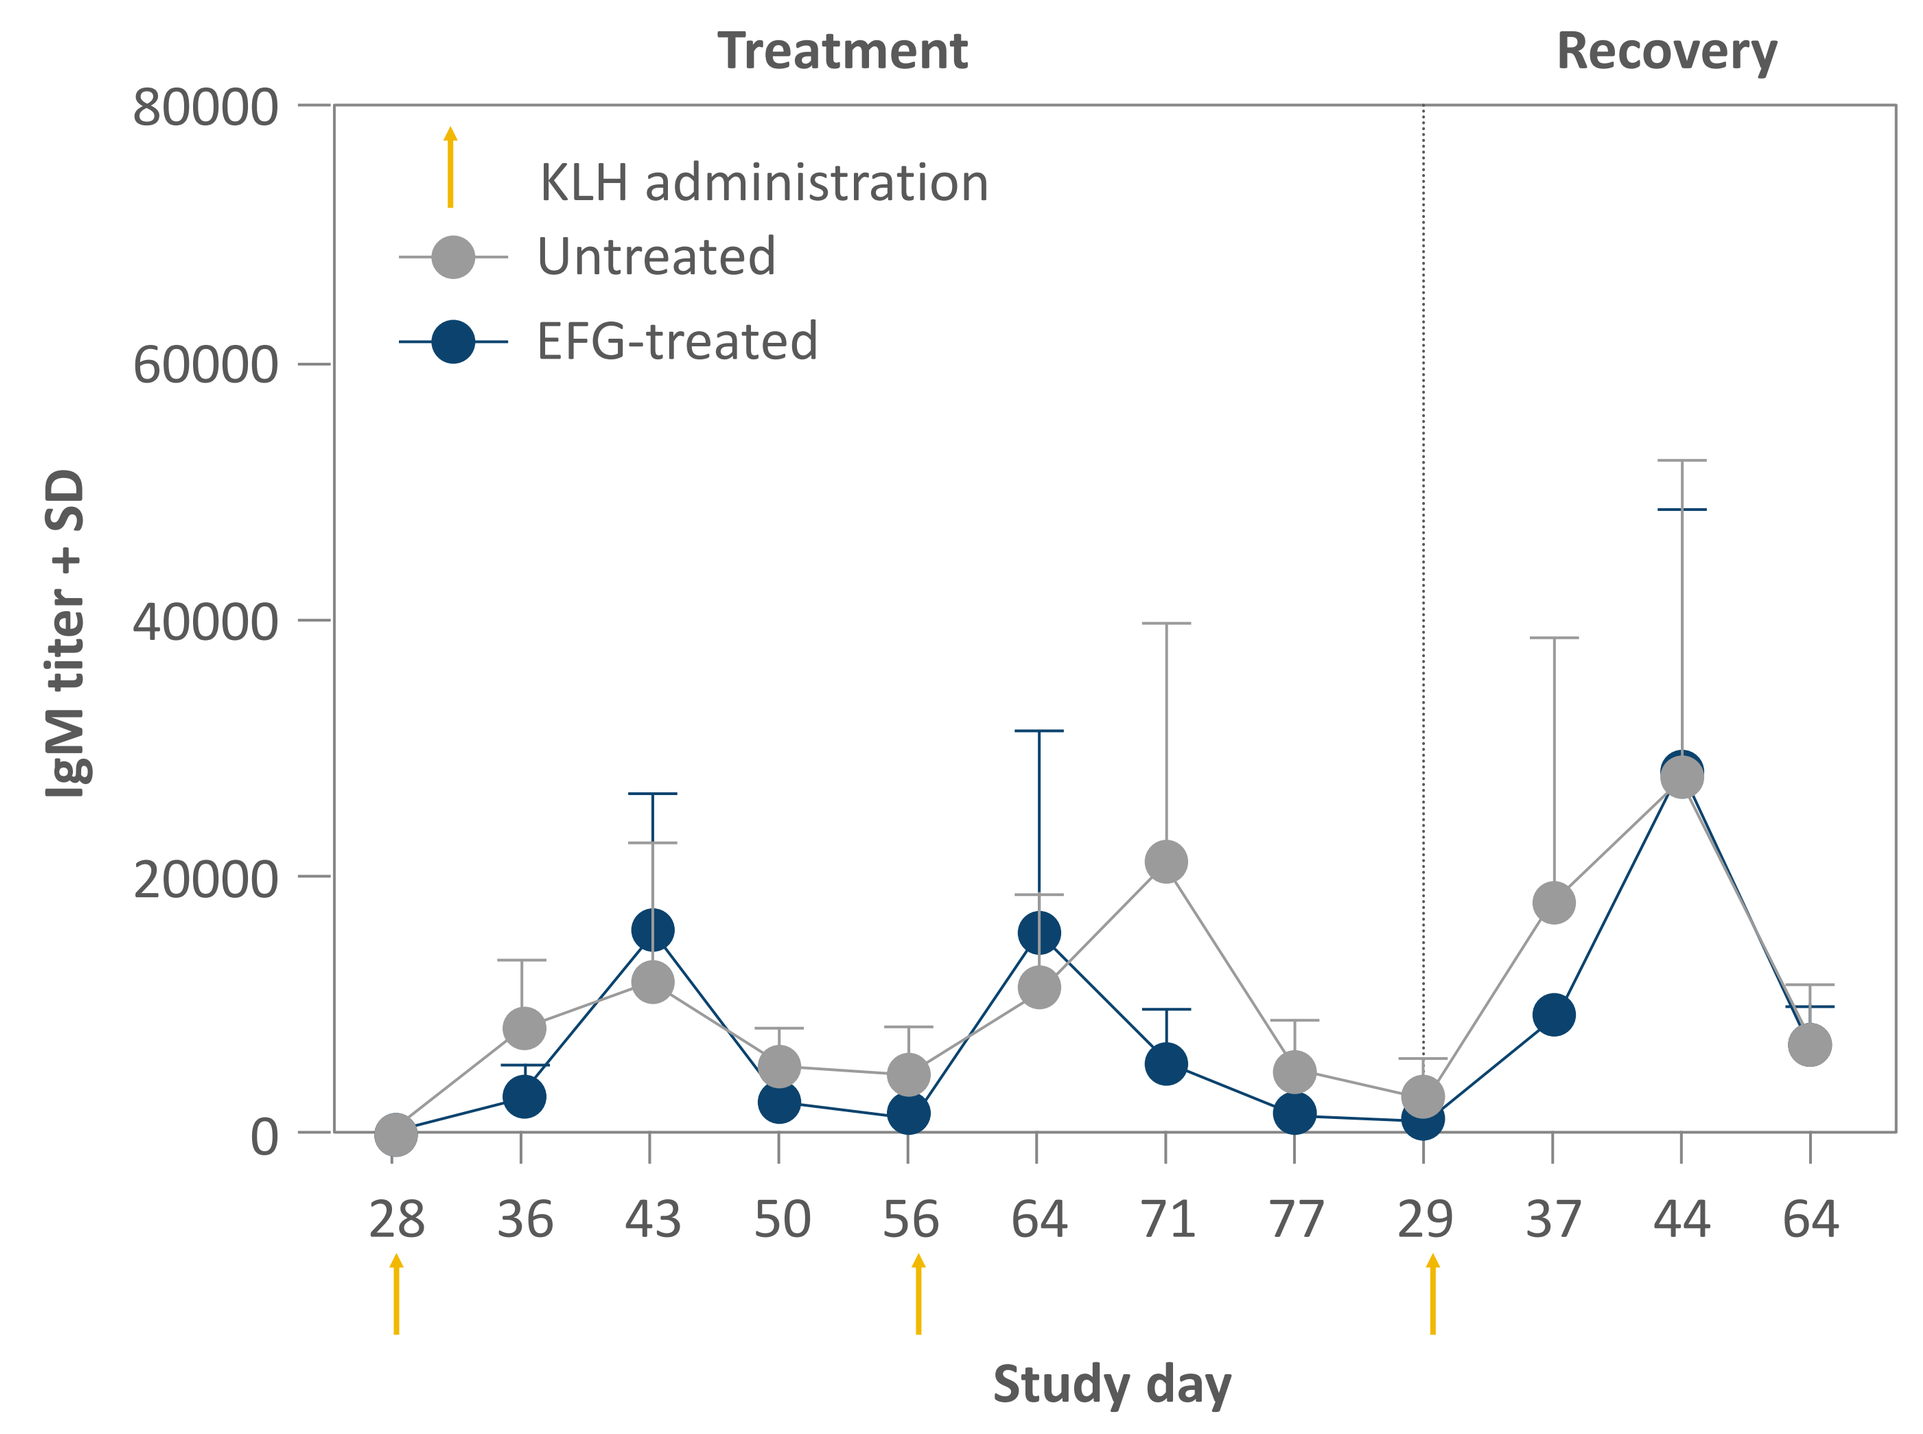

Treatment
Recovery
KLH administration
Untreated
EFG-treated
IgM titer + SD
80000
60000
40000
20000
0
28 36 43 50 56 64 71 77 29 37 44 64
Study day

Cellular Response: KLH Recall

Figure 3. KLH Recall vs Negative Control – Average Male + Female

- Comparable KLH-specific cellular responses between treated vs untreated groups
- No differences between groups during treatment-free period (recovery)

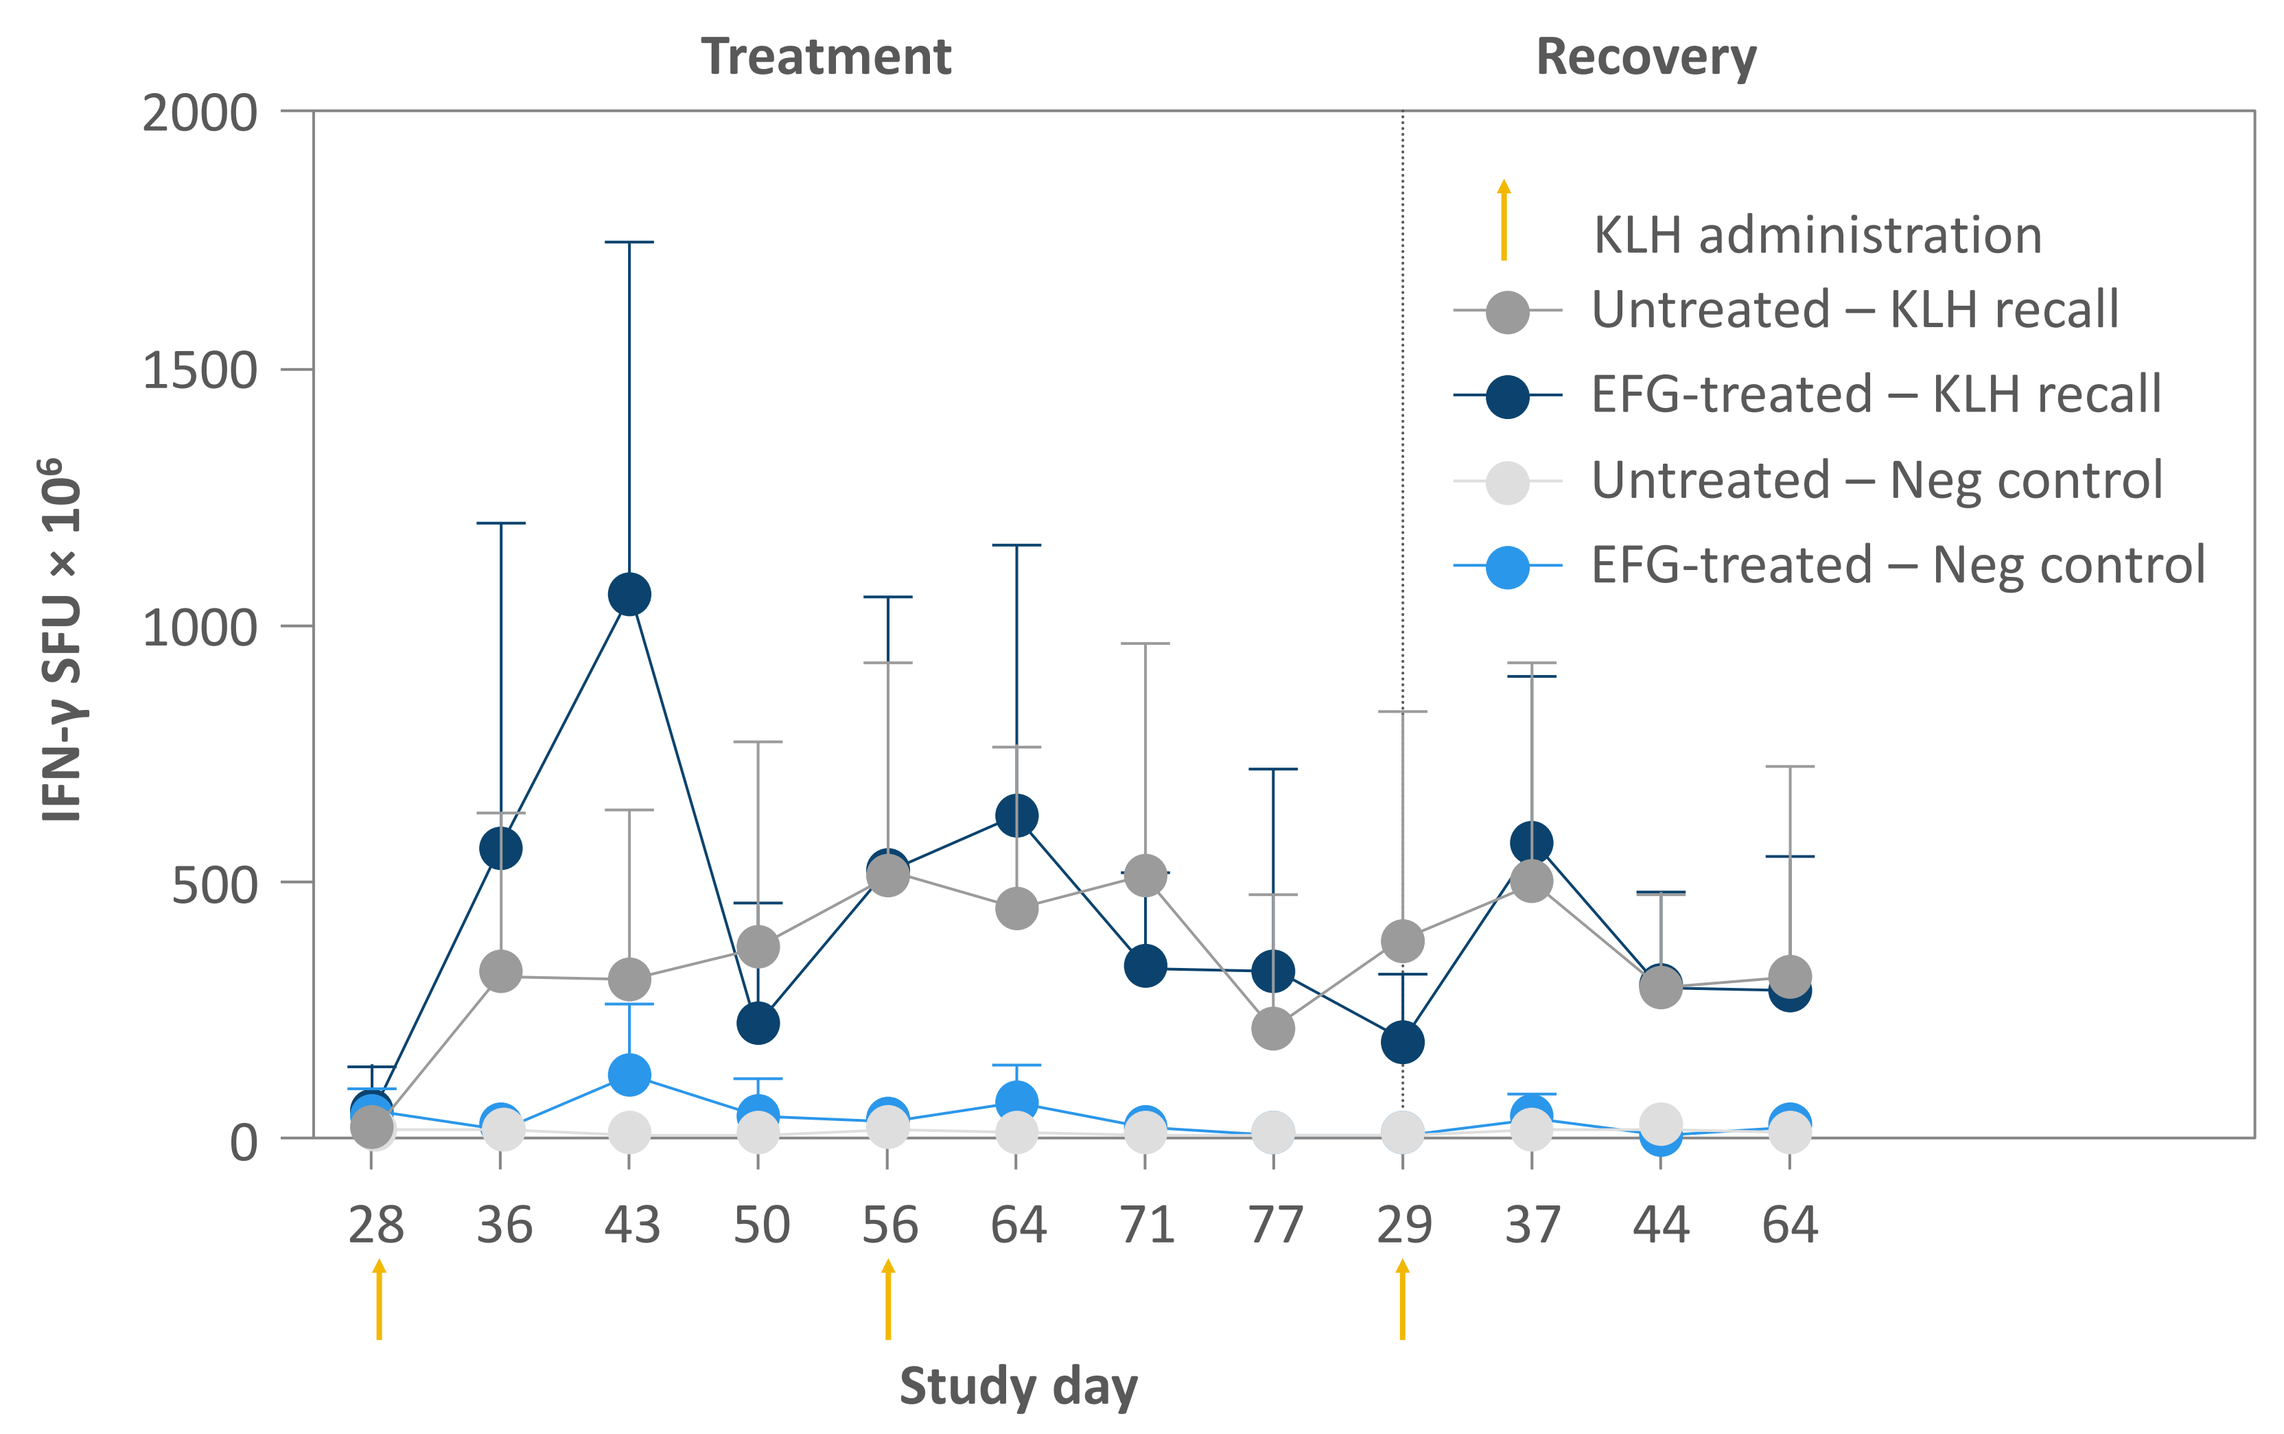

Treatment
Recovery
KLH administration
Untreated – KLH recall
EFG-treated – KLH recall
Untreated – Neg control
EFG-treated – Neg control
IFN-γ SFU × 10⁶
2000
1500
1000
500
0
28 36 43 50 56 64 71 77 29 37 44 64
Study day

Pharmacodynamics: Total IgG Marker

Figure 4. Total IgG (% predose)

- Total IgG titers decreased under efgartigimod treatment, consistent with efgartigimod's expected mechanism of action

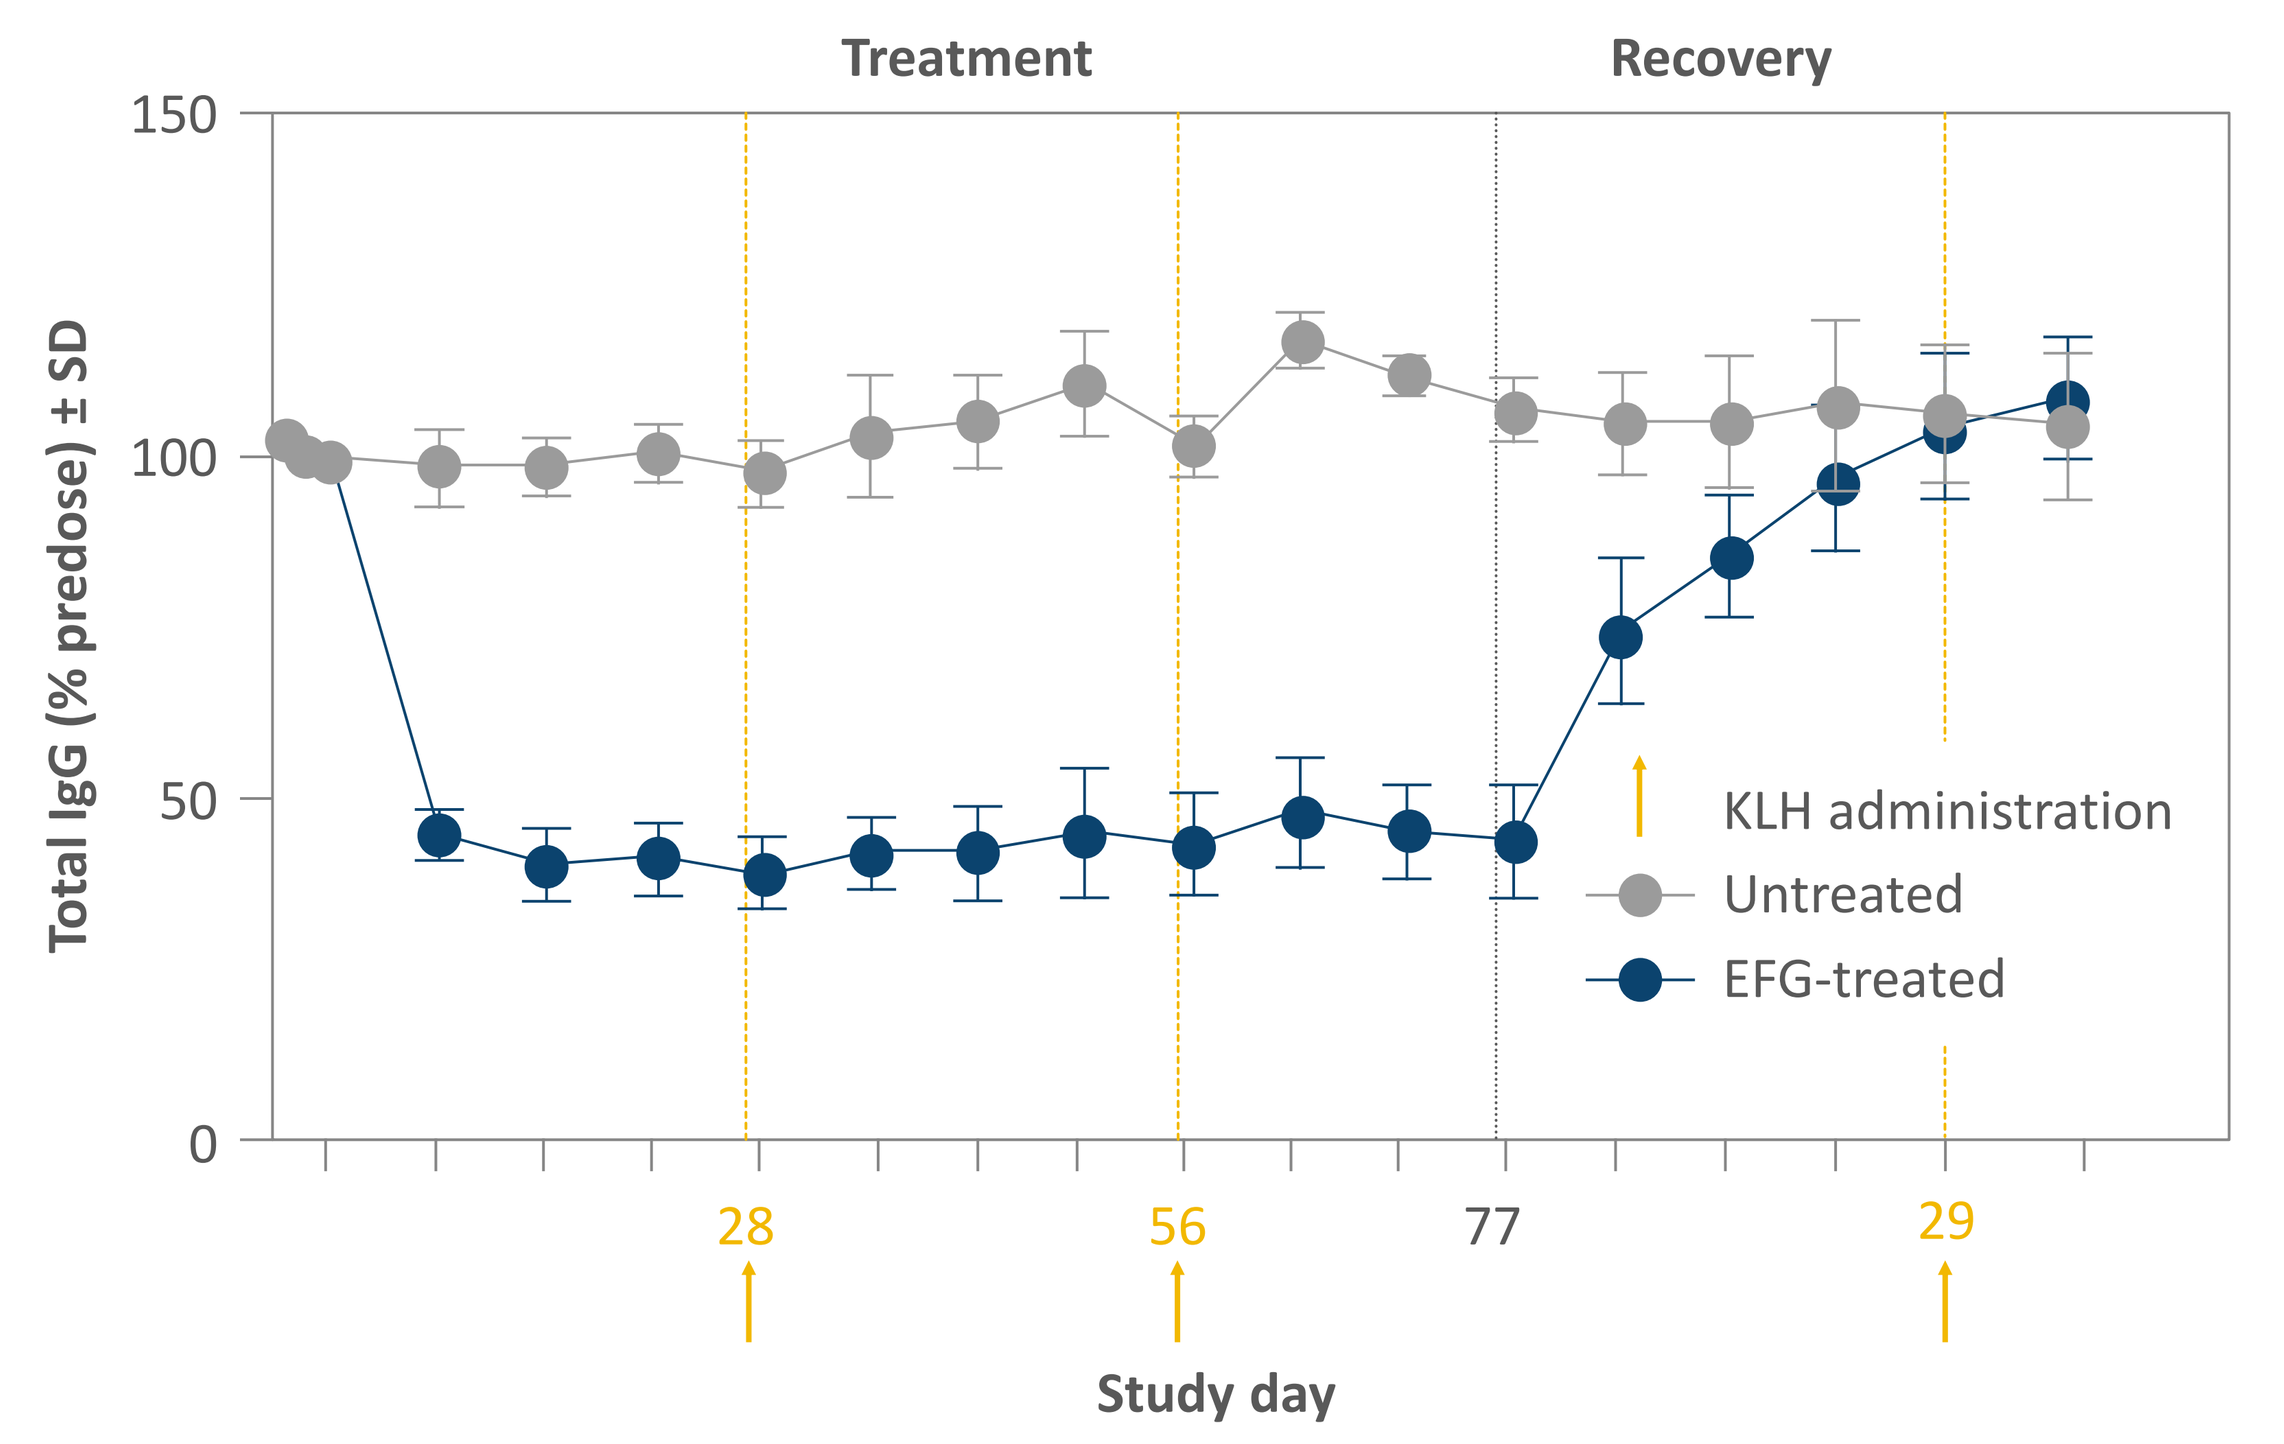

Treatment
Recovery
KLH administration
Untreated
EFG-treated
Total IgG (% predose) ± SD
150
100
50
0
28
56
77
29
Study day

ABBREVIATIONS

COVID-19, coronavirus 2019; EFG, efgartigimod; ELISpot, enzyme-linked immunosorbent spot; FcRn, neonatal Fc receptor; gMG, generalized myasthenia gravis; IFN-γ, interferon-γ; IgG, immunoglobulin G; KLH, keyhole limpet hemocyanin; MMF; mycophenolate mofetil; mRNA, messenger RNA; PBMC, peripheral blood mononuclear cell; SARS-CoV-2, severe acute respiratory syndrome coronavirus 2; SFU, spot-forming unit; TDAR, T-cell–dependent antibody response.

REFERENCES

1. Sesarman A, et al. *Cell Mol Life Sci*. 2010;67(15):2533-2550. **2.** Ulrichts P, et al. *J Clin Invest*. 2018;128(10):4372-4386. **3.** Vaccaro C, et al. *Nat Biotech*. 2005;23(10):1283-1288. **4.** Howard JF Jr, et al. *Lancet Neurol*. 2021;20(7):526-536. **5.** Nixon AE, et al. *Front Immunol.* 2015;6:176. **6.** Ward SE, et al. *Front Immunol*. 2022;13:892534. **7.** Patel SY, et al. *Front Immunol.* 2019;10(33):1-22. **8.** Deepak P, et al. *medRxiv* [Preprint]. 2021;2021.04.05.21254656. **9.** Jolles S, et al. *Clin Exp Immunol.* 2016;188(3):333-341. **10.** Sanders DB, et al. *Neurology.* 2016;87:419-425. **11**. Howard JF Jr, et al. Response to coronavirus 2019 vaccination in patients receiving efgartigimod. Abstract presented at: AANEM 2022; September 21–24; Nashville, TN. **12**. Guptill JT, et al. *Autoimmunity*. 2022;Aug 29;1-12 (Online ahead of print). **13.** Swaminathan A, et al. *Br J Clin Pharmacol*. 2014;78(5):1135-1142. **14**. Picotti JR, et al. *J Immunotoxicol.* 2005;2(4):191-196.

ACKNOWLEDGMENTS AND DISCLOSURES: The authors gratefully acknowledge the trial investigators. **JC, JB, GP, SP, MM, OB, PU, SS, PV, DD,** and **CK** are employees of argenx; **MC, DT, CC,** and **GM** are employees of Labcorp Early Development Laboratories Ltd. Medical writing and editorial support for this presentation was provided by PRECISION Value and Health and funded by argenx.

argenx

Presented at the American Neurological Association (ANA) Annual Meeting; October 22–25, 2022; Chicago, IL, USA