REAL-WORLD USAGE PATTERNS OF VYVGART IN ADULTS WITH GENERALIZED MYASTHENIA GRAVIS IN THE UNITED STATES

Qi C[1]; Brauer E[1]; Gelinas D[1]; Goyal A[2]; Bergin C[1]; Osipa LA[1]; Bhuwalka A[3]; Suthagar T[3]; Phillips G[1]

[1]argenx US Inc., Boston, MA, USA; [2]ZS Associates, Princeton, NJ, USA; [3]ZS Associates, Bengaluru, India

Introduction and Objectives

- Myasthenia gravis (MG) is a rare autoimmune disorder associated with the failure of neuromuscular junction transmission affecting ~60,000 patients in the United States (US).[1,2] Most MG cases progress to generalized MG (gMG) with severe weakness in bulbar, limb, trunk, and respiratory muscles contributing to significant patient burden.[3,4] Although available treatment options for gMG can be effective, they are frequently burdensome, have substantial side effects, or do not alleviate symptoms, leaving considerable unmet needs.[5-7]
- Efgartigimod (VYVGART®) is a human IgG1 Fc fragment, a natural ligand of the neonatal Fc receptor (FcRn) that is a key pathogenic driver in gMG.[7] In the ADAPT phase 3 trial, efgartigimod demonstrated safety, efficacy, and tolerability in patients with gMG and was subsequently approved for use in the US for the treatment of gMG in December 2021.[7-9]
- One unique feature of ADAPT was individualized dosing based on clinical response.[7] After the first treatment cycle (one cycle consisted of 4 infusions, at 1 infusion per week), subsequent cycles were administered according to individual Myasthenia Gravis Activities of Daily Living (MG-ADL) responses during a follow-up of ≥5 weeks.[7] The median time between the last infusion of the 1st cycle and the start of the 2nd cycle was 7 weeks (10 weeks from the start of the 1st cycle), in the ADAPT trial.[7]
- Based on the prescribing information, clinicians in the real world evaluate gMG symptoms following each VYVGART treatment cycle to determine if or when a subsequent treatment cycle should be initiated.[9] Better understanding of real-world VYVGART usage patterns in gMG is critical to augment ADAPT/ADAPT+ data. The objective of this study was to evaluate real-world usage patterns of VYVGART among patients with gMG who initiated VYVGART treatment and enrolled in the My VYVGART Path patient support program. We report our findings up to September 15, 2022, with an additional updated cut on January 23, 2023.

Methods

Initial dataset description and inclusion criteria

- This study used data obtained from My VYVGART Path, a patient support program that provides personalized Nurse Case Manager support for enrolled patients with gMG. Through regular contact, My VYVGART Path Nurse Case Managers capture information including age, gender, weight, and VYVGART treatment schedules of those enrolled.
- Patients with gMG (aged ≥18 years) enrolled in My VYVGART Path who had initiated VYVGART treatment by September 15, 2022, were screened for inclusion in this study. This cohort represents a subset of the real-world population initiating VYVGART. Patients with conflicting or unclear baseline characteristics or demographic information were excluded.
- Of the patients included, the average length of the 1st VYVGART treatment cycle was evaluated using data available from patients who had completed their 1st VYVGART treatment cycle. To evaluate the average time gap between the 1st and 2nd VYVGART treatment cycles, a subset of patients who had completed their 1st VYVGART treatment cycle and initiated their 2nd treatment cycle (i.e., patients whose 4th infusion date of their 1st treatment cycle and 1st infusion date of 2nd treatment cycle were available in the dataset), as of September 15, 2022, were identified for subsequent analyses. An additional analysis using data as of January 23rd, 2022 was also performed.
- The scope of the analysis was restricted to the time period between the 1st and 2nd VYVGART treatment cycles in order to ensure the cohort consisted of a sufficient number of patients, considering that VYVGART was approved for use in the US in December 2021.[8]

Definitions of treatment cycles and gap between treatment cycles (Figure 1)

VYVGART treatment cycle

One VYVGART treatment cycle was defined as an intravenous infusion once per week for 4 consecutive weeks.

Gap between 2 treatment cycles

The duration between the last infusion of one cycle and the 1st infusion of the subsequent cycle.

Figure 1. Definition of treatment cycles and gap between treatment cycles

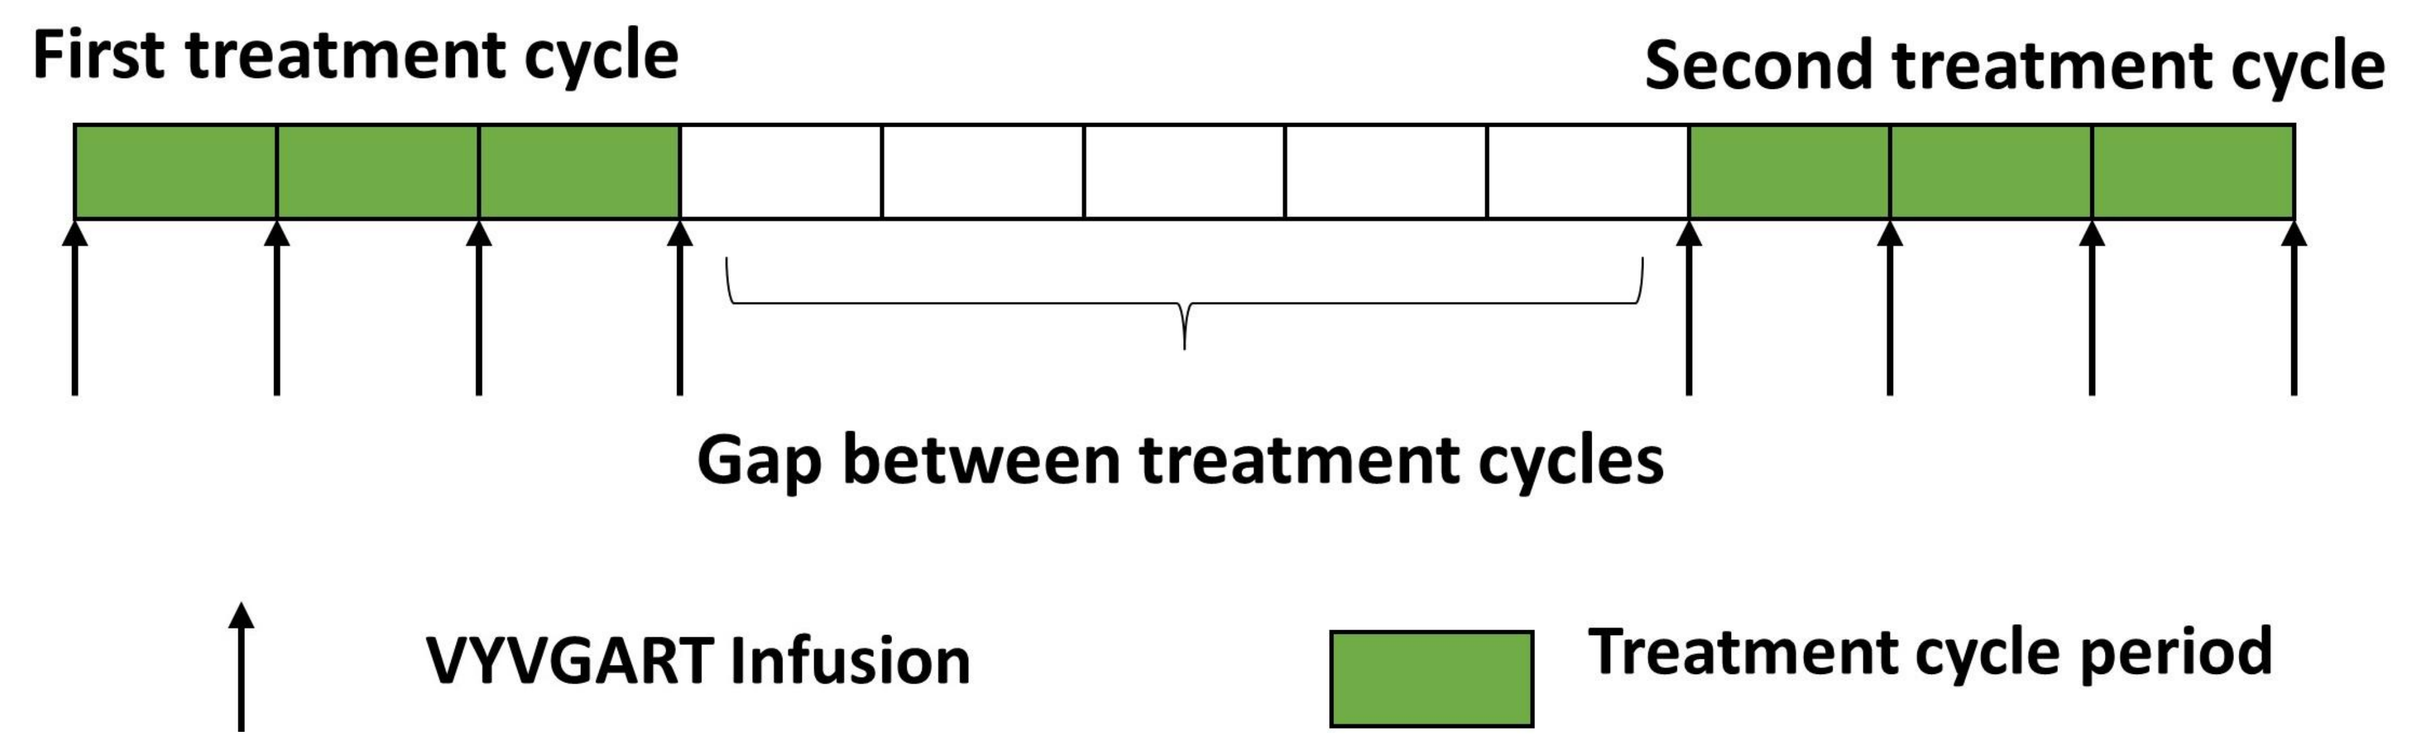
First treatment cycle
Second treatment cycle
Gap between treatment cycles
VYVGART Infusion
Treatment cycle period

Results

1. Study population and baseline characteristics

- 1,399 patients who were enrolled in the My VYVGART Path program and had initiated VYVGART treatment before September 15, 2022, were identified and included in the study.

Figure 2. Baseline patient demographics and characteristics (N=1,399)

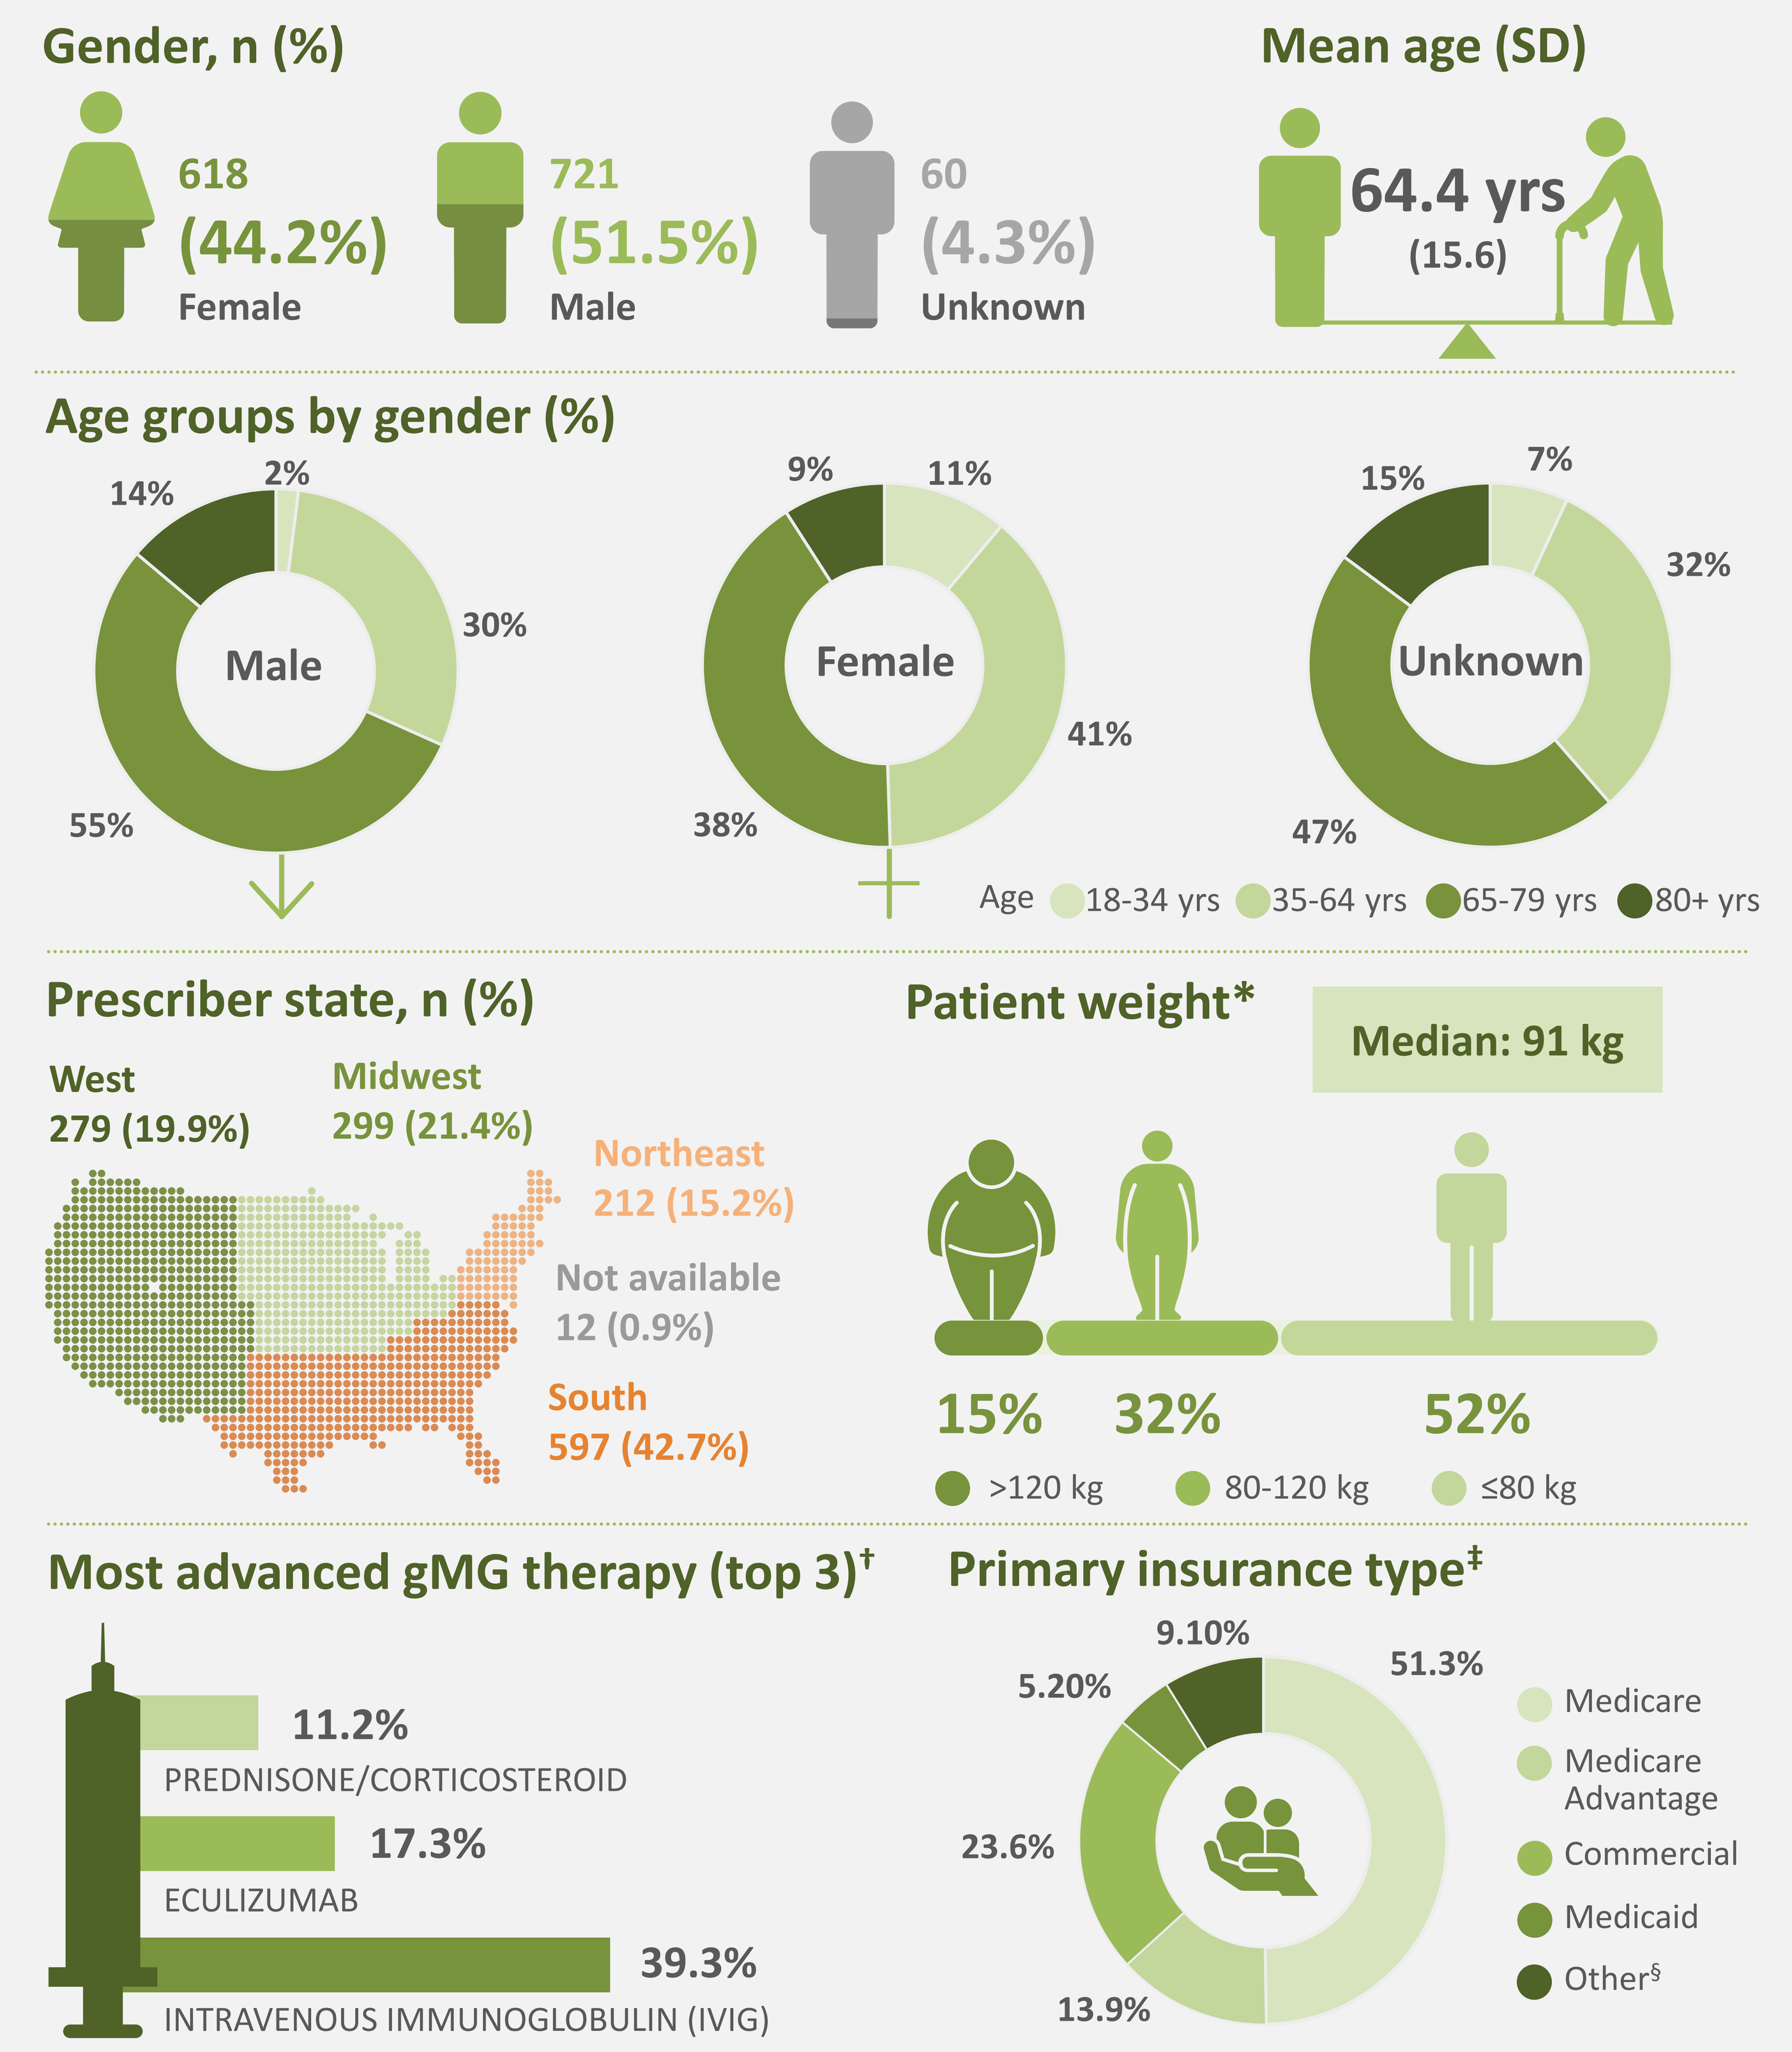
Gender, n (%)
618 (44.2%) Female
721 (51.5%) Male
60 (4.3%) Unknown
Mean age (SD)
64.4 yrs (15.6)
Age groups by gender (%)
Male: 2% 30% 55% 14%
Female: 11% 41% 38% 9%
Unknown: 7% 32% 47% 15%
Age 18-34 yrs 35-64 yrs 65-79 yrs 80+ yrs
Prescriber state, n (%)
West 279 (19.9%)
Midwest 299 (21.4%)
Northeast 212 (15.2%)
Not available 12 (0.9%)
South 597 (42.7%)
Patient weight*
Median: 91 kg
15% 32% 52%
>120 kg 80-120 kg ≤80 kg
Most advanced gMG therapy (top 3)†
11.2% PREDNISONE/CORTICOSTEROID
17.3% ECULIZUMAB
39.3% INTRAVENOUS IMMUNOGLOBULIN (IVIG)
Primary insurance type‡
51.3% 13.9% 23.6% 5.20% 9.10%
Medicare
Medicare Advantage
Commercial
Medicaid
Other§

SD, standard deviation. *Weight information was missing for 68 of 1,399 patients. †The most advanced gMG therapy each patient had received before VYVGART was identified using the following priority order: THYMECTOMY < MESTINON/ANTI-ACETYLCHOLINESTERASE INHIBITOR < PREDNISONE/CORTICOSTEROID < NSIST < IVIG < RITUXIMAB < ECULIZUMAB < PLEX. For example, if a patient record showed prior use of corticosteroids, NSISTs, and IVIg, IVIg was recorded at the most advanced therapy based on the priority order. Only the top 3 most common categories are shown in the figure, with the full results below. Previous gMG therapy information was not reported for 268 patients (19%). ‡Percentages may not add up to 100% as patients may have had multiple plan subscriptions during the analysis duration. §Other included government, patient assistance programs, federal employee programs, and Veteran's Affairs/Department of Defense.

- A higher proportion of VYVGART initiators identified were males, compared with females. The most common age group among males was 65–79 years, while the most common age group among females was 35–64 years. VYVGART prescribers were most frequently located in the Southern United States **(Figure 2)**.
- The most common advanced gMG therapy among the cohort was intravenous immunoglobulin (IVIg) (39%), followed by eculizumab (17%), prednisone/corticosteroid (11%), non-steroidal immunosuppressing treatments (NSISTs) (6%), rituximab (5%), anti-acetylcholinesterase inhibitors (1%), and plasma exchange (PLEX) (1%) **(Figure 2)**.

2. Length of 1st VYVGART treatment cycle

- Among patients who had completed their 1st VYVGART treatment cycle (n=988 of 1,399), the average time over which the 4 infusions of the 1st treatment cycle were administered was 21.5 days.
 - 83.2% (n=822) completed their 1st treatment cycle within 21 days, and 98.2% (n=970) between 14 and 28 days.

3. Gap between the 1st and 2nd VYVGART treatment cycles

- Of the 1,399 patients who had initiated VYVGART treatment, 440 patients (31.5%) had completed their 1st VYVGART treatment cycle and initiated the 2nd treatment cycle as of September 15, 2022 **(Figure 3)**.

Figure 3. Patient selection

1,399
Patients with treatment cycle record in dataset as of September 15, 2022

440
Patients who completed their 1st treatment cycle and initiated their 2nd treatment cycle as of September 15, 2022

Figure 4. Distribution of time gap between 1st and 2nd VYVGART treatment cycles (n=440)

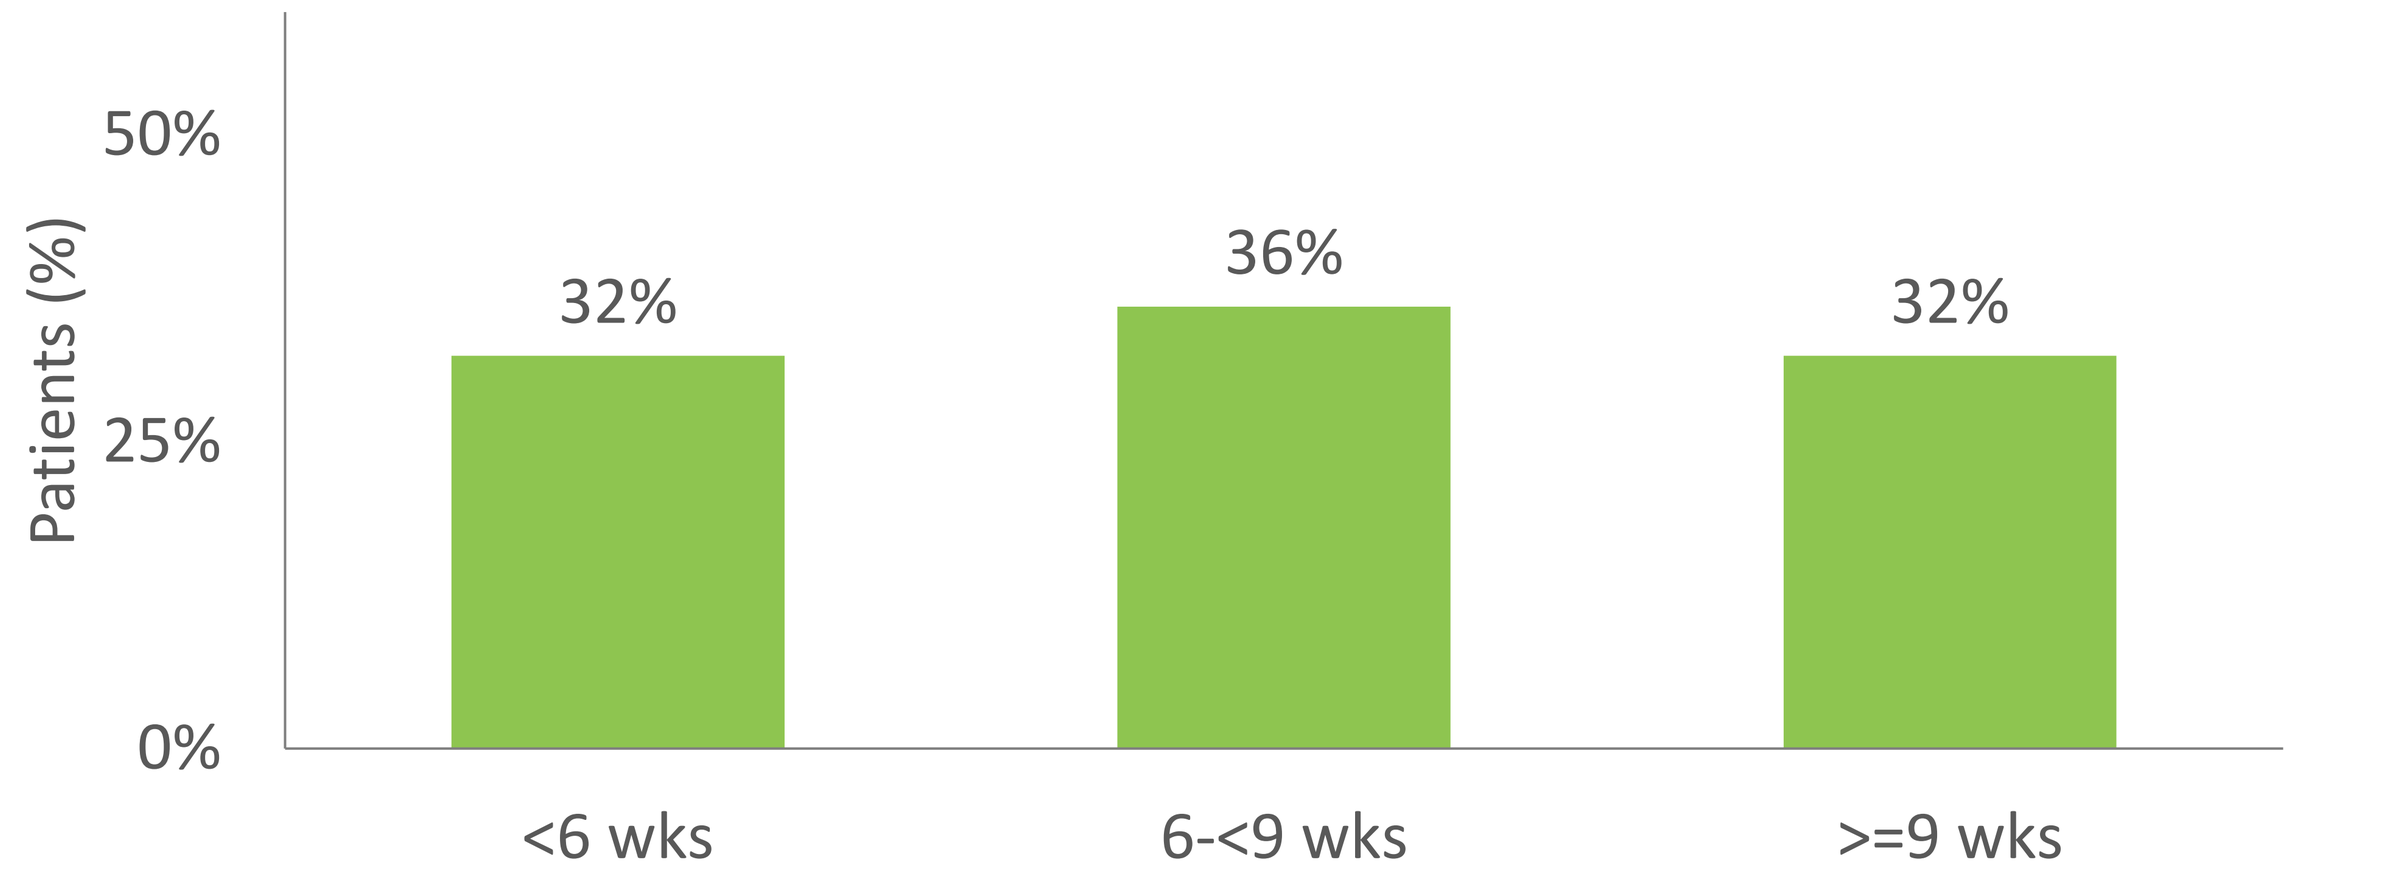
Patients (%)
50%
25%
0%
32%
36%
32%
<6 wks
6-<9 wks
>=9 wks

- For patients who initiated their 2nd VYVGART treatment cycle (n=440), the gap between the last infusion of the 1st cycle and the first infusion of the 2nd cycle was <6 weeks for 32.3% of patients, between 6 and <9 weeks for 35.7% of patients, and ≥9 weeks for 32.0% of patients **(Figure 4)**.
- The average gap between the last infusion of the 1st treatment cycle and the first infusion of the 2nd cycle was 50.7 days, and the median gap was 48.0 days.
- For validation, a separate analysis was conducted using actual dispense dates reported in specialty pharmacy (SP)-reported dispense data. For patients whose data could be identified in both SP and My VYVGART Path datasets, the gap reported between the 1st and 2nd cycles was consistent across both analyses.

Additional updated analysis

Updated data: Gap between the 1st and 2nd VYVGART treatment cycles

- Utilizing the same study design and methodology, we updated the analysis incorporating more recent data considering all patients who had initiated VYVGART treatment on or before **January 23, 2023**.
- As of **January 23, 2023,** 2,215 patients had initiated VYVGART treatment, and 1,108 (50%) had completed their 1st VYVGART treatment cycle and initiated the 2nd treatment cycle. Among these 1,108 patients:
 - The gap between the last infusion of the 1st cycle and the 1st infusion of the 2nd cycle was <6 weeks for 32.3% of patients, between 6 and <9 weeks for 35.7% of patients, and ≥9 weeks for 32.0% of patients **(Figure 5)**.
 - The average and median gap between the 1st and 2nd treatment cycles was 58.0 days and 50.0 days, respectively.
- The utilization patterns remain similar with more data captured. The slight increase in the gap between 1st and 2nd treatment cycles is expected, as the initial analysis sample would have comprised more frequent VYVGART users.

Figure 5. Distribution of time gap between 1st and 2nd VYVGART treatment cycles, updated cohort (n=1,108)

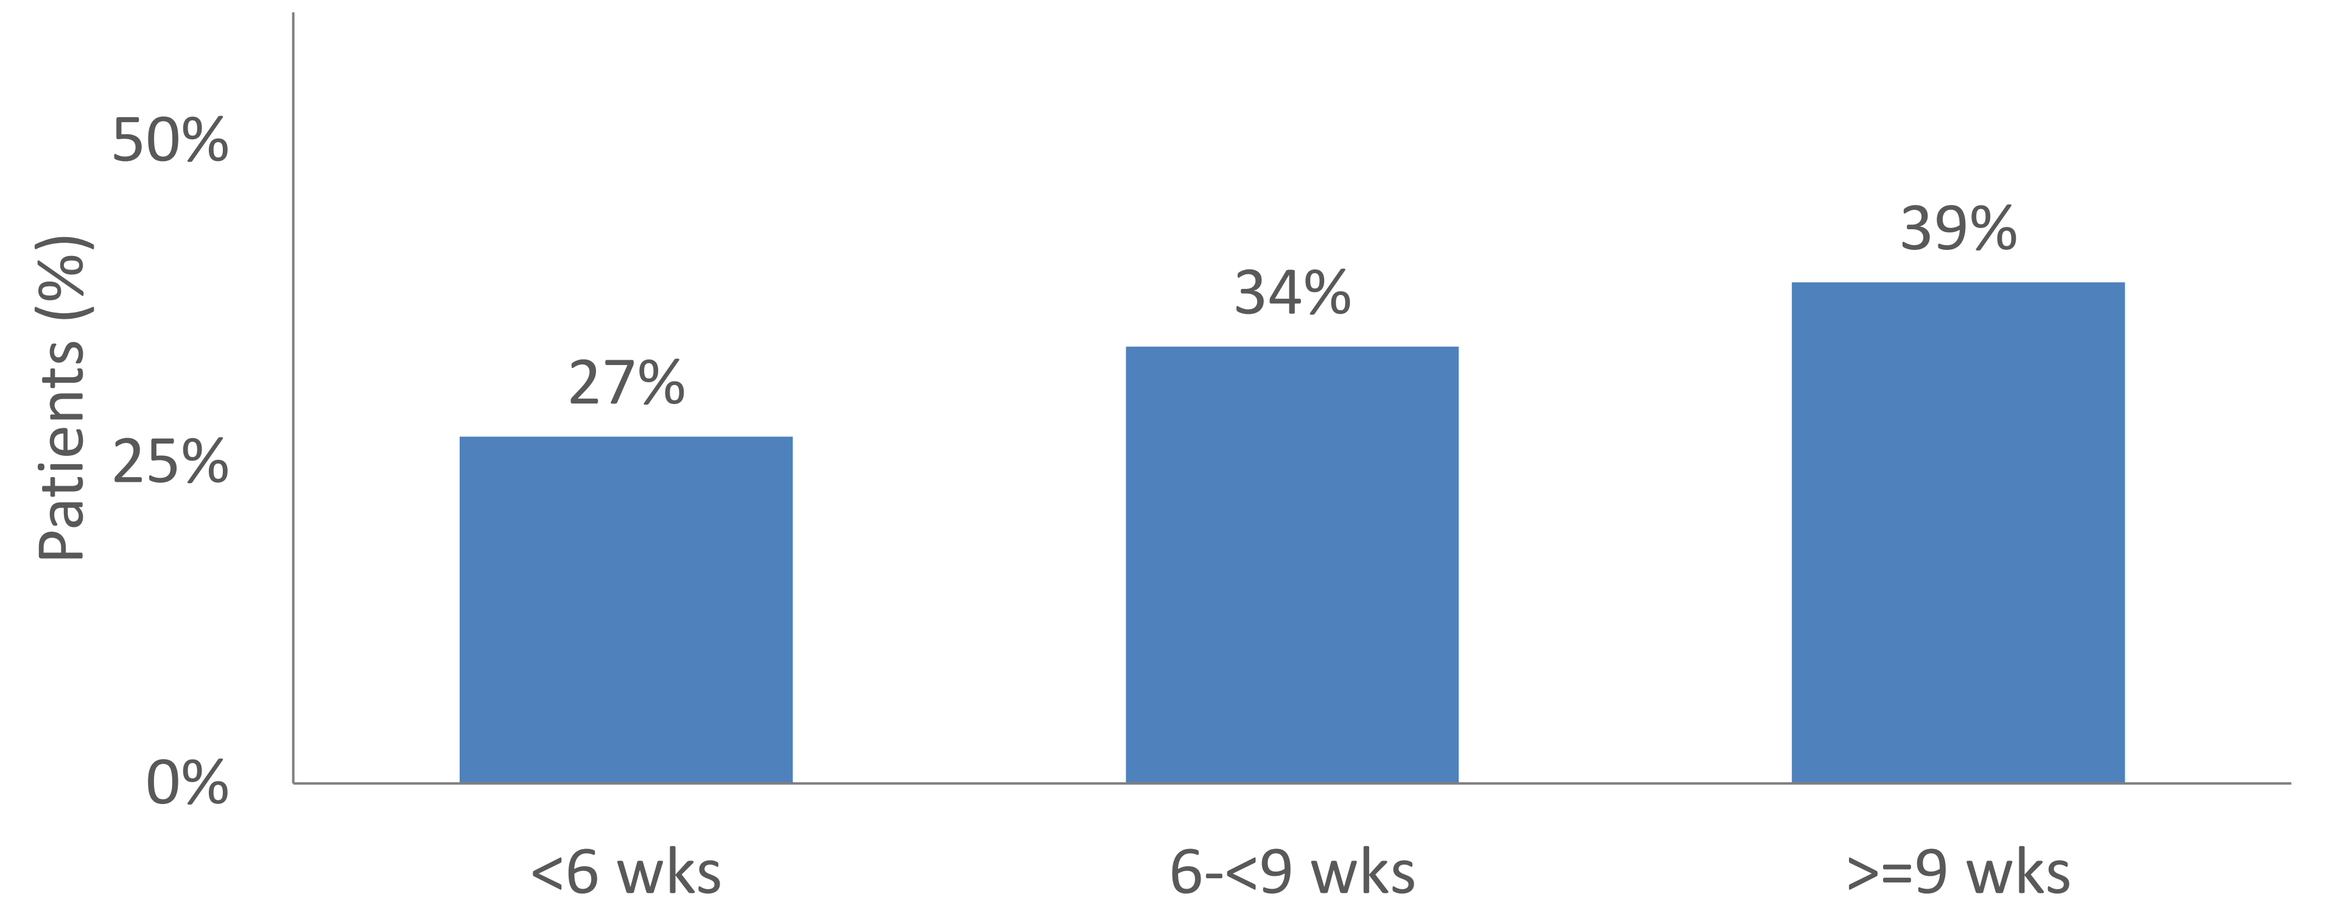
Patients (%)
50%
25%
0%
27%
34%
39%
<6 wks
6-<9 wks
>=9 wks

Discussions and Conclusions

- Among 1,399 patients with gMG in the US who initiated VYVGART treatment and were enrolled in the My VYVGART Path program as of **September 15, 2022**, 440 initiated the 2nd treatment cycle. The average gap between the last infusion of the 1st cycle and 1st infusion of the 2nd cycle was 50.7 days (median was 48 days). These real-world estimates of the duration are consistent with the results reported in the ADAPT trial (7 weeks).[10] The results remain consistent with the updated data cut including 1,108 patients (data as of **January 23, 2023**).
- Some limitations of this study should be considered, including that the data analyzed were self-reported by patients who were contacted by My VYVGART Path Nurse Case Managers who recorded the data. Additionally, the study population only included patients enrolled in the My VYVGART Path program in the US who had initiated VYVGART by September 15, 2022 (or January 23, 2023, in the additional updated analysis), after VYVGART became approved for use in December 2021.
- Nevertheless, our results represent the most comprehensive and up-to-date real-world utilization patterns of VYVGART based on a dataset with direct insights into infusion dates and treatment cycle structures. Consistent results were observed in an objective specialty pharmacy-reported dispense dataset, supporting the robustness of our analysis.
- Our results suggest that early real-world utilization patterns of VYVGART are consistent with ADAPT trial results, but these patterns are expected to continue to shift over a longer follow-up period. Future studies should continue to gather real-world data from a larger, increasingly diverse population of patients with gMG to help optimize outcomes as the gMG treatment landscape continues to rapidly evolve.

Financial support This study was funded by argenx US, Inc. (Boston, MA, USA).

Disclosures and acknowledgements The material in this poster has not been previously presented or published. CQ, EB, DG, CB, LAO, and GP are employees of argenx. AG, AB, and TS are employees of ZS Associates and serve as paid consultants for argenx. Medical writing and editorial support was provided by Mai Sato, PhD (ZS Associates) and Katherine Gora (ZS Associates) in accordance with Good Publication Practice (GPP) 2022 guidelines and was funded by argenx.

References

1. Conti-Fine BM, et al. J Clin Invest. Nov 2006;116(11):2843-2854. 2. Phillips LH. Semin Neurol. Mar 2004;24(1):17-20. 3. Jani-Acsadi A, Lisak RP. J Neurol Sci. Oct 15 2007;261(1-2):127-133. 4. Phillips G, et al. Front Neurol. 2021;12:809999. 5. Bacci ED, et al. BMC Neurol. Dec 21 2019;19(1):335. 6. Dalakas MC. Curr Opin Neurol. Oct 2020;33(5):545-552. 7. Howard JF, Jr., et al. Lancet Neurol. Jul 2021;20(7):526-536. 8. The US Food and Drug Administration. FDA Approves New Treatment for Myasthenia Gravis. FDA News Release. Accessed Jan 20, 2023. https://www.fda.gov/news-events/press-announcements/fda-approves-new-treatment-myasthenia-gravis. 9. argenx BV. VYVGART™ (efgartigimod alfa-fcab) [package insert]. U.S. Food and Drug Administration. Accessed Jan 19, 2023. https://www.accessdata.fda.gov/drugsatfda_docs/label/2021/761195s000lbl.pdf.

45th Annual Carrell-Krusen Neuromuscular Symposium | February 23–24, 2023 | Dallas, TX