Poster #134

Efgartigimod Demonstrates Consistent Improvements in Generalized Myasthenia Gravis Across Patient Subgroups, Including Early in Diagnosis

Elissa Ritt,[1] Vera Bril,[2,3] Tuan Vu,[4] Edward Brauer,[1] René Kerstens,[1] James F. Howard Jr[5]; in collaboration with the ADAPT Investigator Study Group

[1]argenx, Ghent, Belgium; [2]Ellen & Martin Prosserman Centre for Neuromuscular Diseases, University Health Network, Toronto, Ontario, Canada; [3]University of Toronto, Toronto, Ontario, Canada; [4]Department of Neurology, University of South Florida, Morsani College of Medicine, Tampa, FL, USA; [5]Department of Neurology, The University of North Carolina, Chapel Hill, NC, USA

INTRODUCTION

Efgartigimod Mechanism of Action: Blocking FcRn

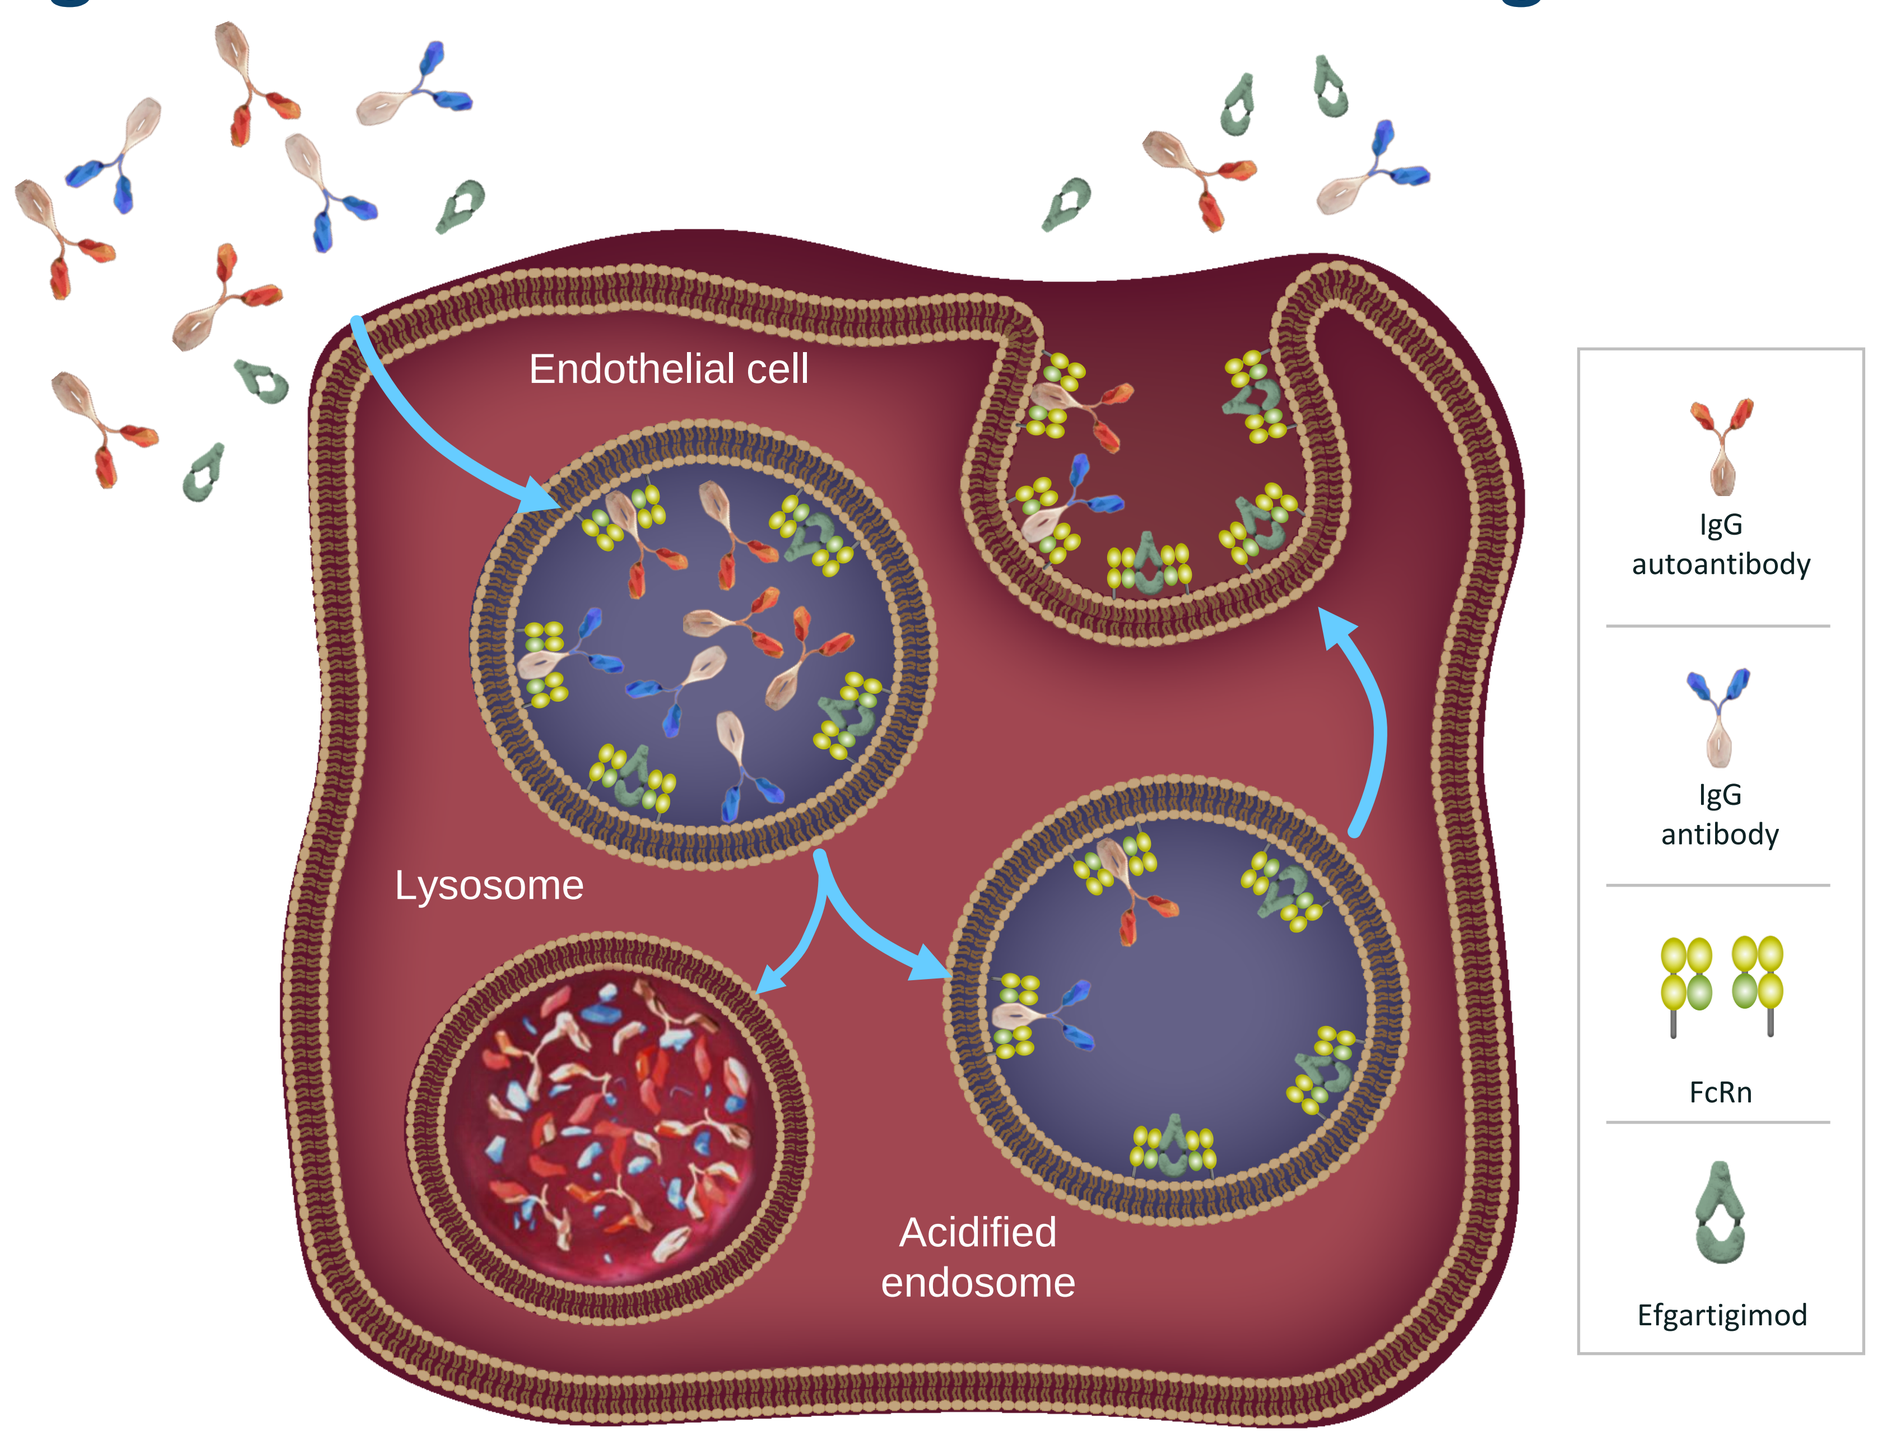

Endothelial cell
Lysosome
Acidified endosome
IgG autoantibody
IgG antibody
FcRn
Efgartigimod

- FcRn recycles IgG, extending its half-life and maintaining serum concentration[1]
- Efgartigimod is a human IgG1 Fc fragment, a natural ligand of FcRn, engineered for increased affinity to FcRn[2,3]
- Efgartigimod was designed to outcompete endogenous IgG, preventing recycling and promoting IgG lysosomal degradation without directly impacting its production[2-6]
 - Targeted reduction of all IgG subtypes
 - No impact on IgM or IgA
 - No reduction in albumin levels
 - No increase in cholesterol

SUMMARY

- Efgartigimod demonstrates consistent improvements compared to placebo, regardless of disease duration, concomitant therapy, prior therapies (including thymectomy), or prior treatment failures
- These data suggest that efgartigimod is an effective treatment in a broad patient population, including early in disease, and early in the treatment journey of patients with gMG
- Efgartigimod was well tolerated, with most adverse events being mild or moderate in severity

METHODS

ADAPT was a 26-week, global, multicenter, randomized, double-blind, placebo-controlled, phase 3 trial evaluating the safety and efficacy of efgartigimod in patients with gMG[4]

- A post hoc analysis of data collected from AChR-Ab+ patients in ADAPT was performed in subgroups based on baseline disease factors and concomitant gMG therapies

adapt myasthenia gravis study

ADAPT
Patients randomized 1:1 to receive cycles of 4 infusions at weekly intervals of 10 mg/kg IV efgartigimod or placebo[4]

Entry criteria
- MGFA class II, III, IV
- AChR-Ab positive or negative
- MG-ADL score ≥5 (>50% nonocular)
- On ≥1 stable gMG treatment[a]
- IgG ≥6 g/L

Efgartigimod n=84
1:1
Placebo n=83

26 weeks (≤3 cycles[b])

Initiation of new treatment cycle:
- ≥5 weeks between cycles
- MG-ADL score ≥5[c]
- MG-ADL score within 2 points of baseline

Primary Endpoint:
- Percentage of AChR-Ab+ patients who were **MG-ADL responders** after cycle 1, **defined by a ≥2-point reduction from cycle 1 baseline score for ≥4 consecutive weeks**, with the first decrease occurring ≤1 week after last study drug infusion

Key Secondary Endpoint
- Percentage of AChR-Ab+ patients who were **QMG responders** after cycle 1, **defined by a ≥3-point reduction from cycle 1 baseline score for ≥4 consecutive weeks**, with the first decrease occurring ≤1 week after last study drug infusion

N=151 → **ADAPT+ (Open-Label Extension)**

Note: Beige rectangles within arrow indicates day of efgartigimod infusion. AChR-Ab, acetylcholine receptor antibody; gMG, generalized myasthenia gravis; IgG, immunoglobulin G; IV, intravenous; gMG, generalized myasthenia gravis; MG-ADL, Myasthenia Gravis Activities of Daily Living; MGFA, Myasthenia Gravis Foundation of America; QMG, Quantitative Myasthenia Gravis. [a]Acetylcholinesterase inhibitor, steroid +/or nonsteroidal immunosuppressive therapy. Patients could not change concomitant therapies in ADAPT. [b]≤3 cycles dosed at ≥8 weeks after initial cycle. [c]With >50% from nonocular items.

RESULTS

Table 1. Baseline Characteristics
AChR-Ab+ Patients

	Efgartigimod (n=65)	Placebo (n=64)
Age, mean, y (SD)	44.7 (15.0)	49.2 (15.5)
Sex, female, n (%)	46 (70.8)	40 (62.5)
Time since diagnosis, mean, y (SD)	9.68 (8.3)	8.93 (8.2)
MG-ADL score, mean (SD)	9.0 (2.5)	8.6 (2.1)
QMG score, mean (SD)	16.0 (5.1)	15.2 (4.4)
MGFA class at screening, n (%)		
Class II	28 (43.1)	25 (39.1)
Class III	35 (53.8)	36 (56.3)
Class IV	2 (3.1)	3 (4.7)
Prior treatment with NSIST, n (%)	47 (72.3)	43 (67.2)
MG therapies at baseline, n (%)		
Any NSIST	40 (61.5)	37 (57.8)
Any steroid	46 (70.8)	51 (79.7)
Steroid and NSIST	34 (52.3)	31 (48.4)
AChEI only	13 (20.0)	6 (9.4)

Table 2. Safety Data, Overall Population

	Efgartigimod (n=84)	Placebo (n=83)
AEs,[a] n (%)	65 (77.4)	70 (84.3)
SAEs, n (%)	4 (4.8)	7 (8.4)
Discontinued due to AEs,[b] n (%)	3 (3.6)	3 (3.6)

[a]Most AEs were mild to moderate in severity. [b]Patients treated with efgartigimod: gMG worsening, rectal adenocarcinoma, thrombocytosis (determined to be unlikely related to efgartigimod by the investigator); patients treated with placebo: myocardial ischemia, atrial fibrillation, spinal ligament ossification.

Figure 1. Proportion of MG-ADL and QMG Responders by Disease Duration
AChR-Ab+ Patients, Cycle 1

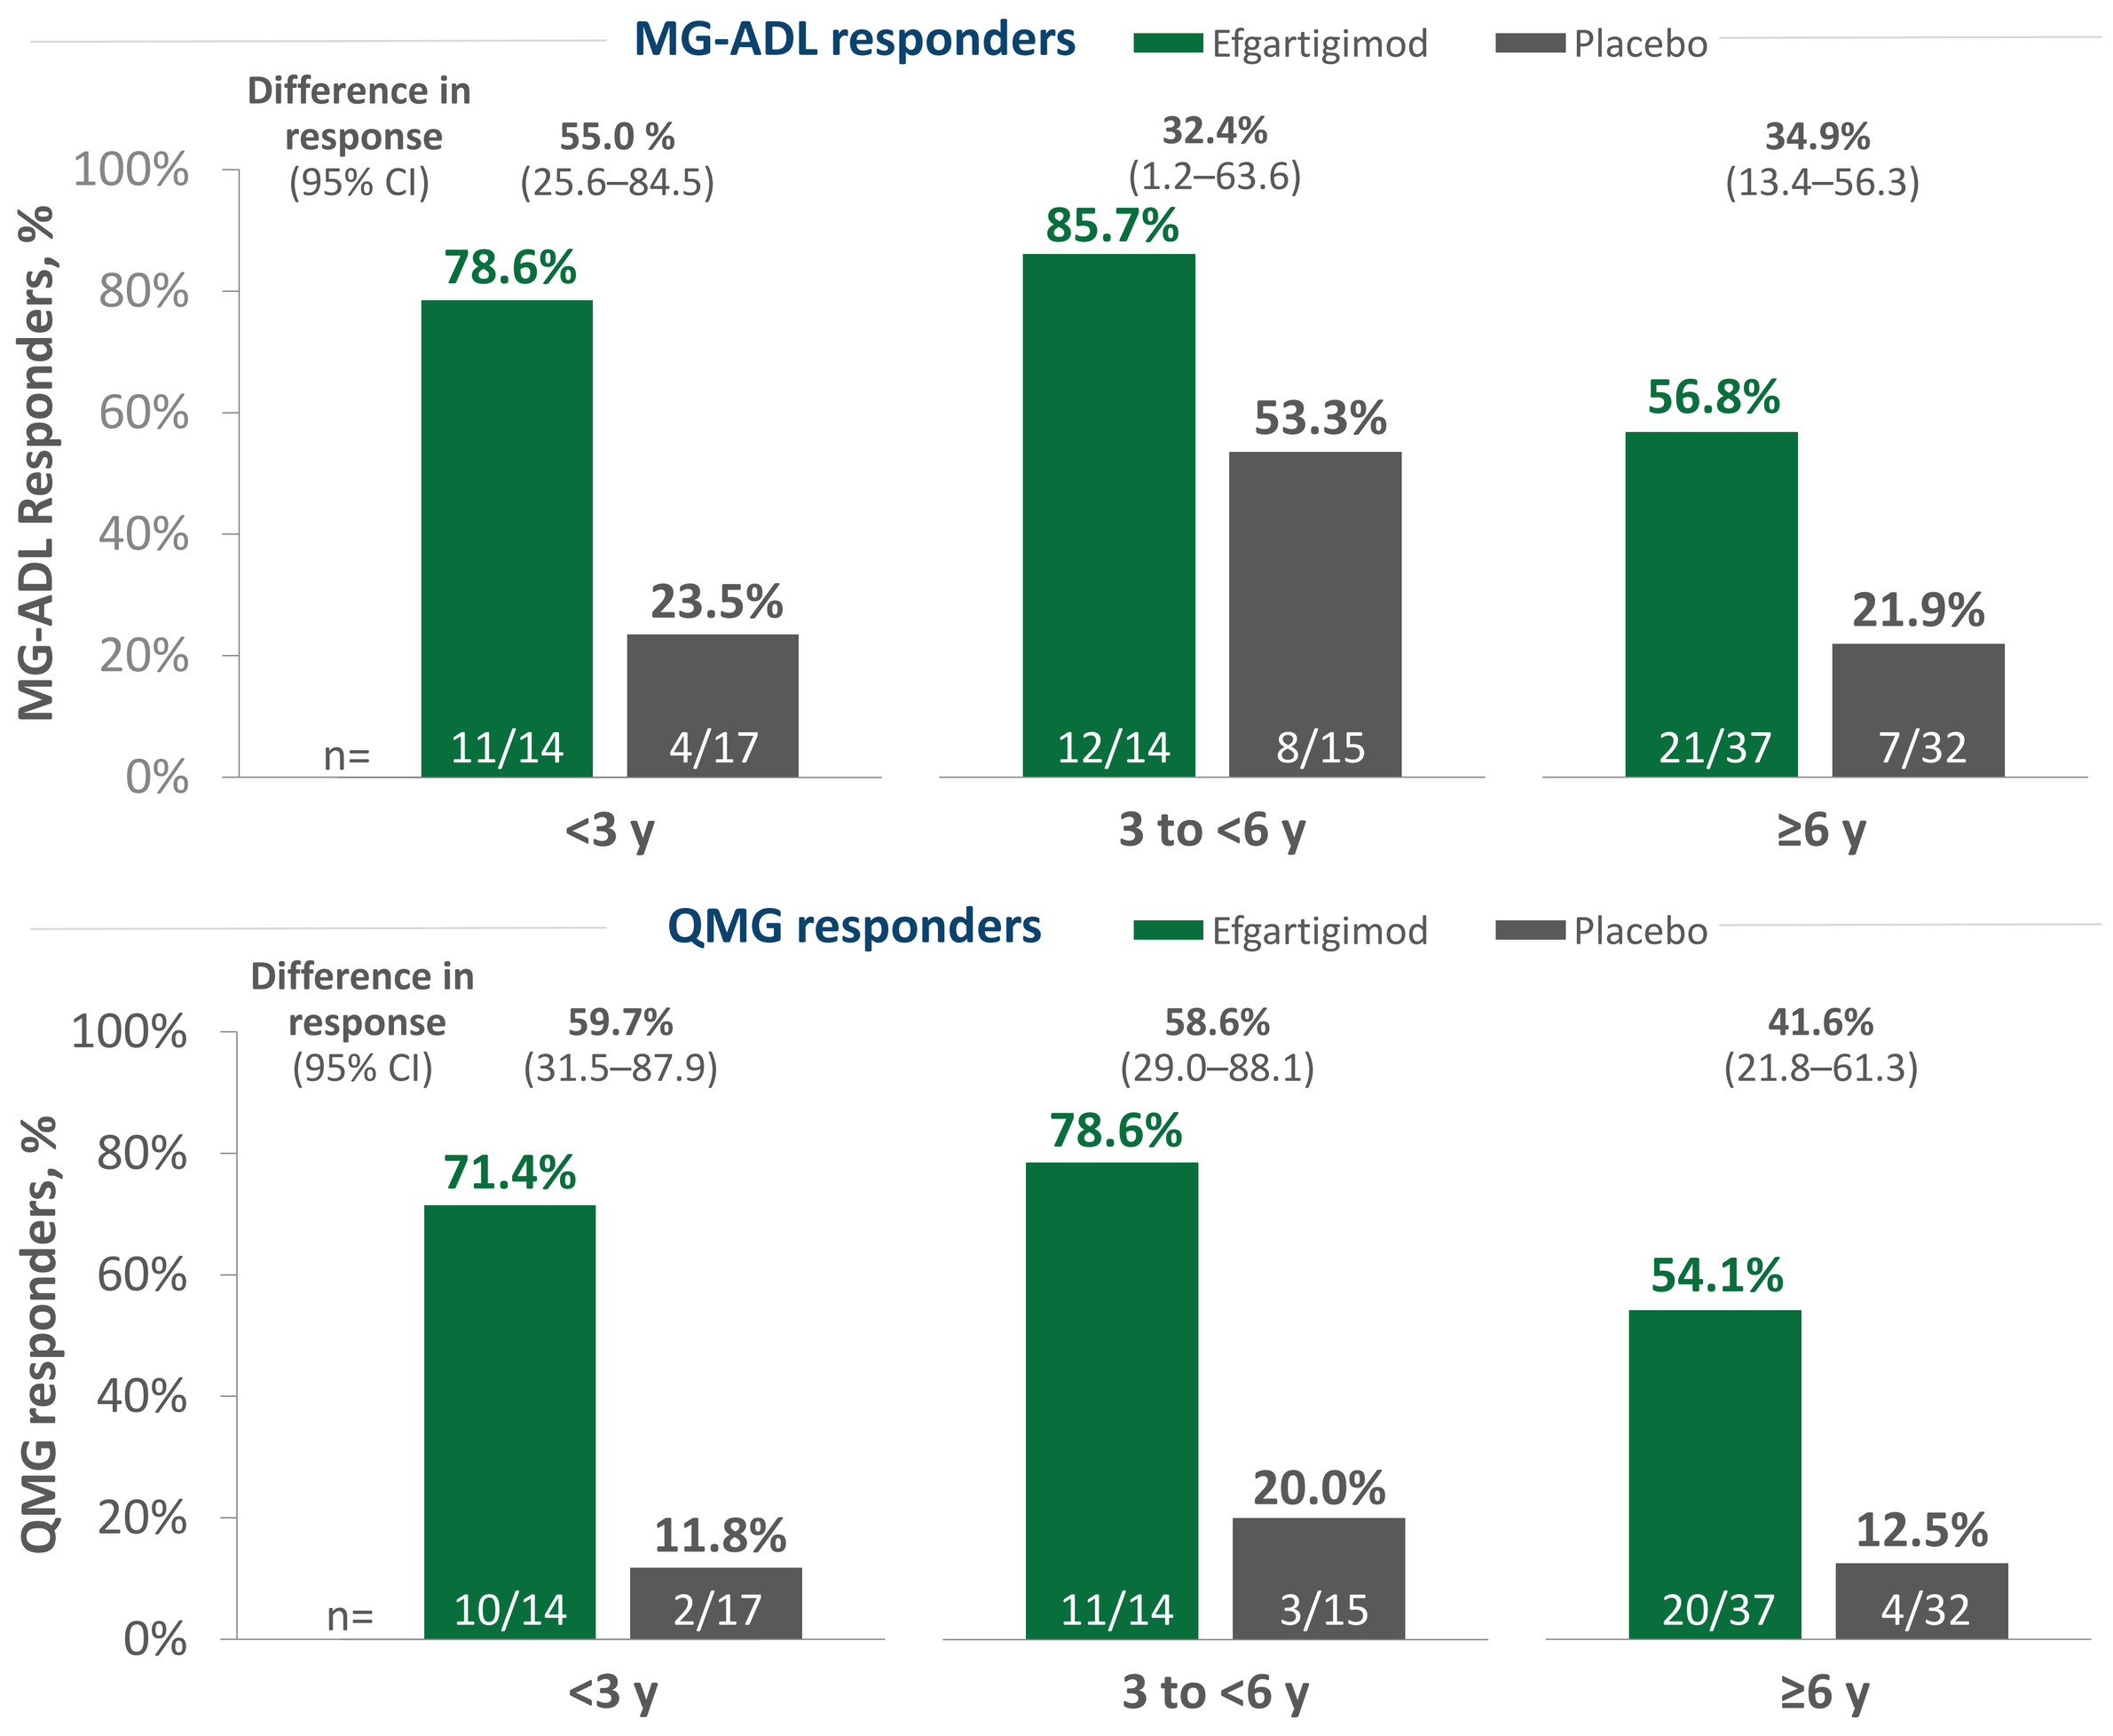

MG-ADL responders
Efgartigimod
Placebo
Difference in response (95% CI)
55.0 % (25.6–84.5)
32.4% (1.2–63.6)
34.9% (13.4–56.3)
MG-ADL Responders, %
78.6%
23.5%
85.7%
53.3%
56.8%
21.9%
n= 11/14 4/17
12/14 8/15
21/37 7/32
<3 y
3 to <6 y
≥6 y
QMG responders
59.7% (31.5–87.9)
58.6% (29.0–88.1)
41.6% (21.8–61.3)
QMG responders, %
71.4%
11.8%
78.6%
20.0%
54.1%
12.5%
n= 10/14 2/17
11/14 3/15
20/37 4/32

Figure 2. Proportion of MG-ADL and QMG Responders by Concomitant Therapies
AChR-Ab+ Patients, Cycle 1

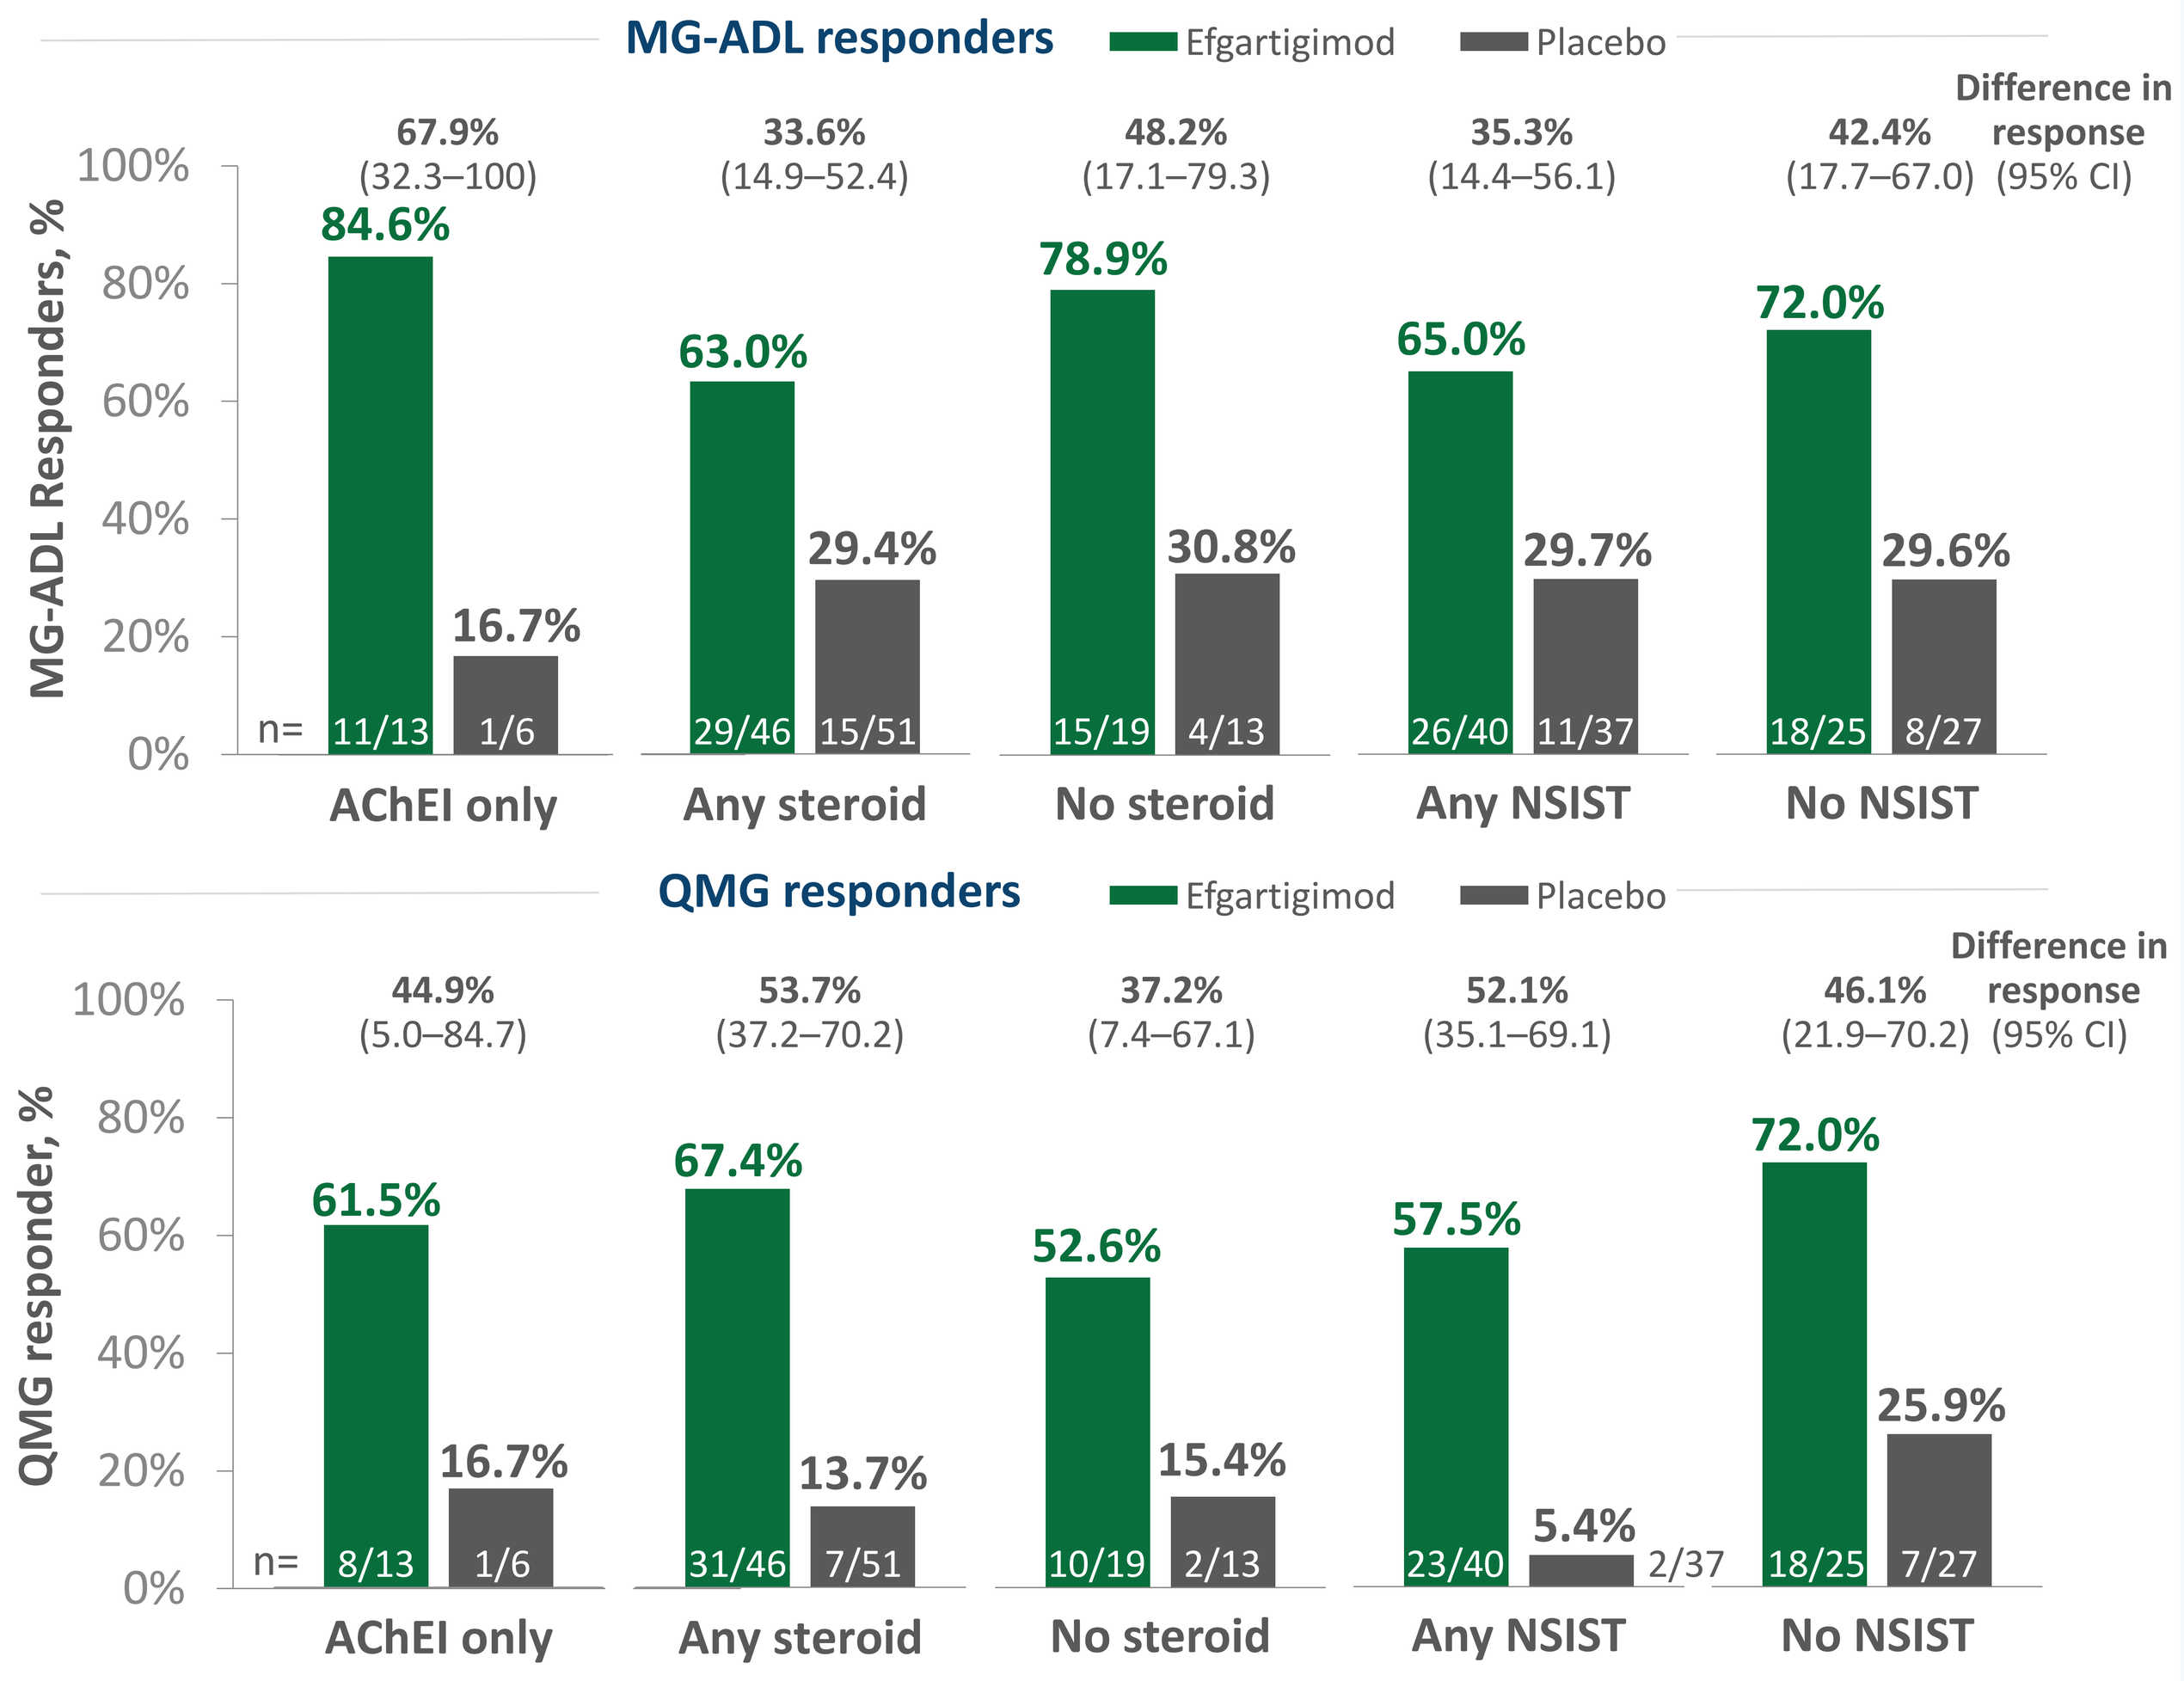

MG-ADL responders
Efgartigimod
Placebo
Difference in response (95% CI)
67.9% (32.3–100)
33.6% (14.9–52.4)
48.2% (17.1–79.3)
35.3% (14.4–56.1)
42.4% (17.7–67.0)
MG-ADL Responders, %
84.6%
16.7%
63.0%
29.4%
78.9%
30.8%
65.0%
29.7%
72.0%
29.6%
n= 11/13 1/6
29/46 15/51
15/19 4/13
26/40 11/37
18/25 8/27
AChEI only
Any steroid
No steroid
Any NSIST
No NSIST
QMG responders
44.9% (5.0–84.7)
53.7% (37.2–70.2)
37.2% (7.4–67.1)
52.1% (35.1–69.1)
46.1% (21.9–70.2)
QMG responder, %
61.5%
16.7%
67.4%
13.7%
52.6%
15.4%
57.5%
5.4%
72.0%
25.9%
n= 8/13 1/6
31/46 7/51
10/19 2/13
23/40 2/37
18/25 7/27

Figure 3. Proportion of MG-ADL and QMG Responders by Prior Therapies
AChR-Ab+ Patients, Cycle 1

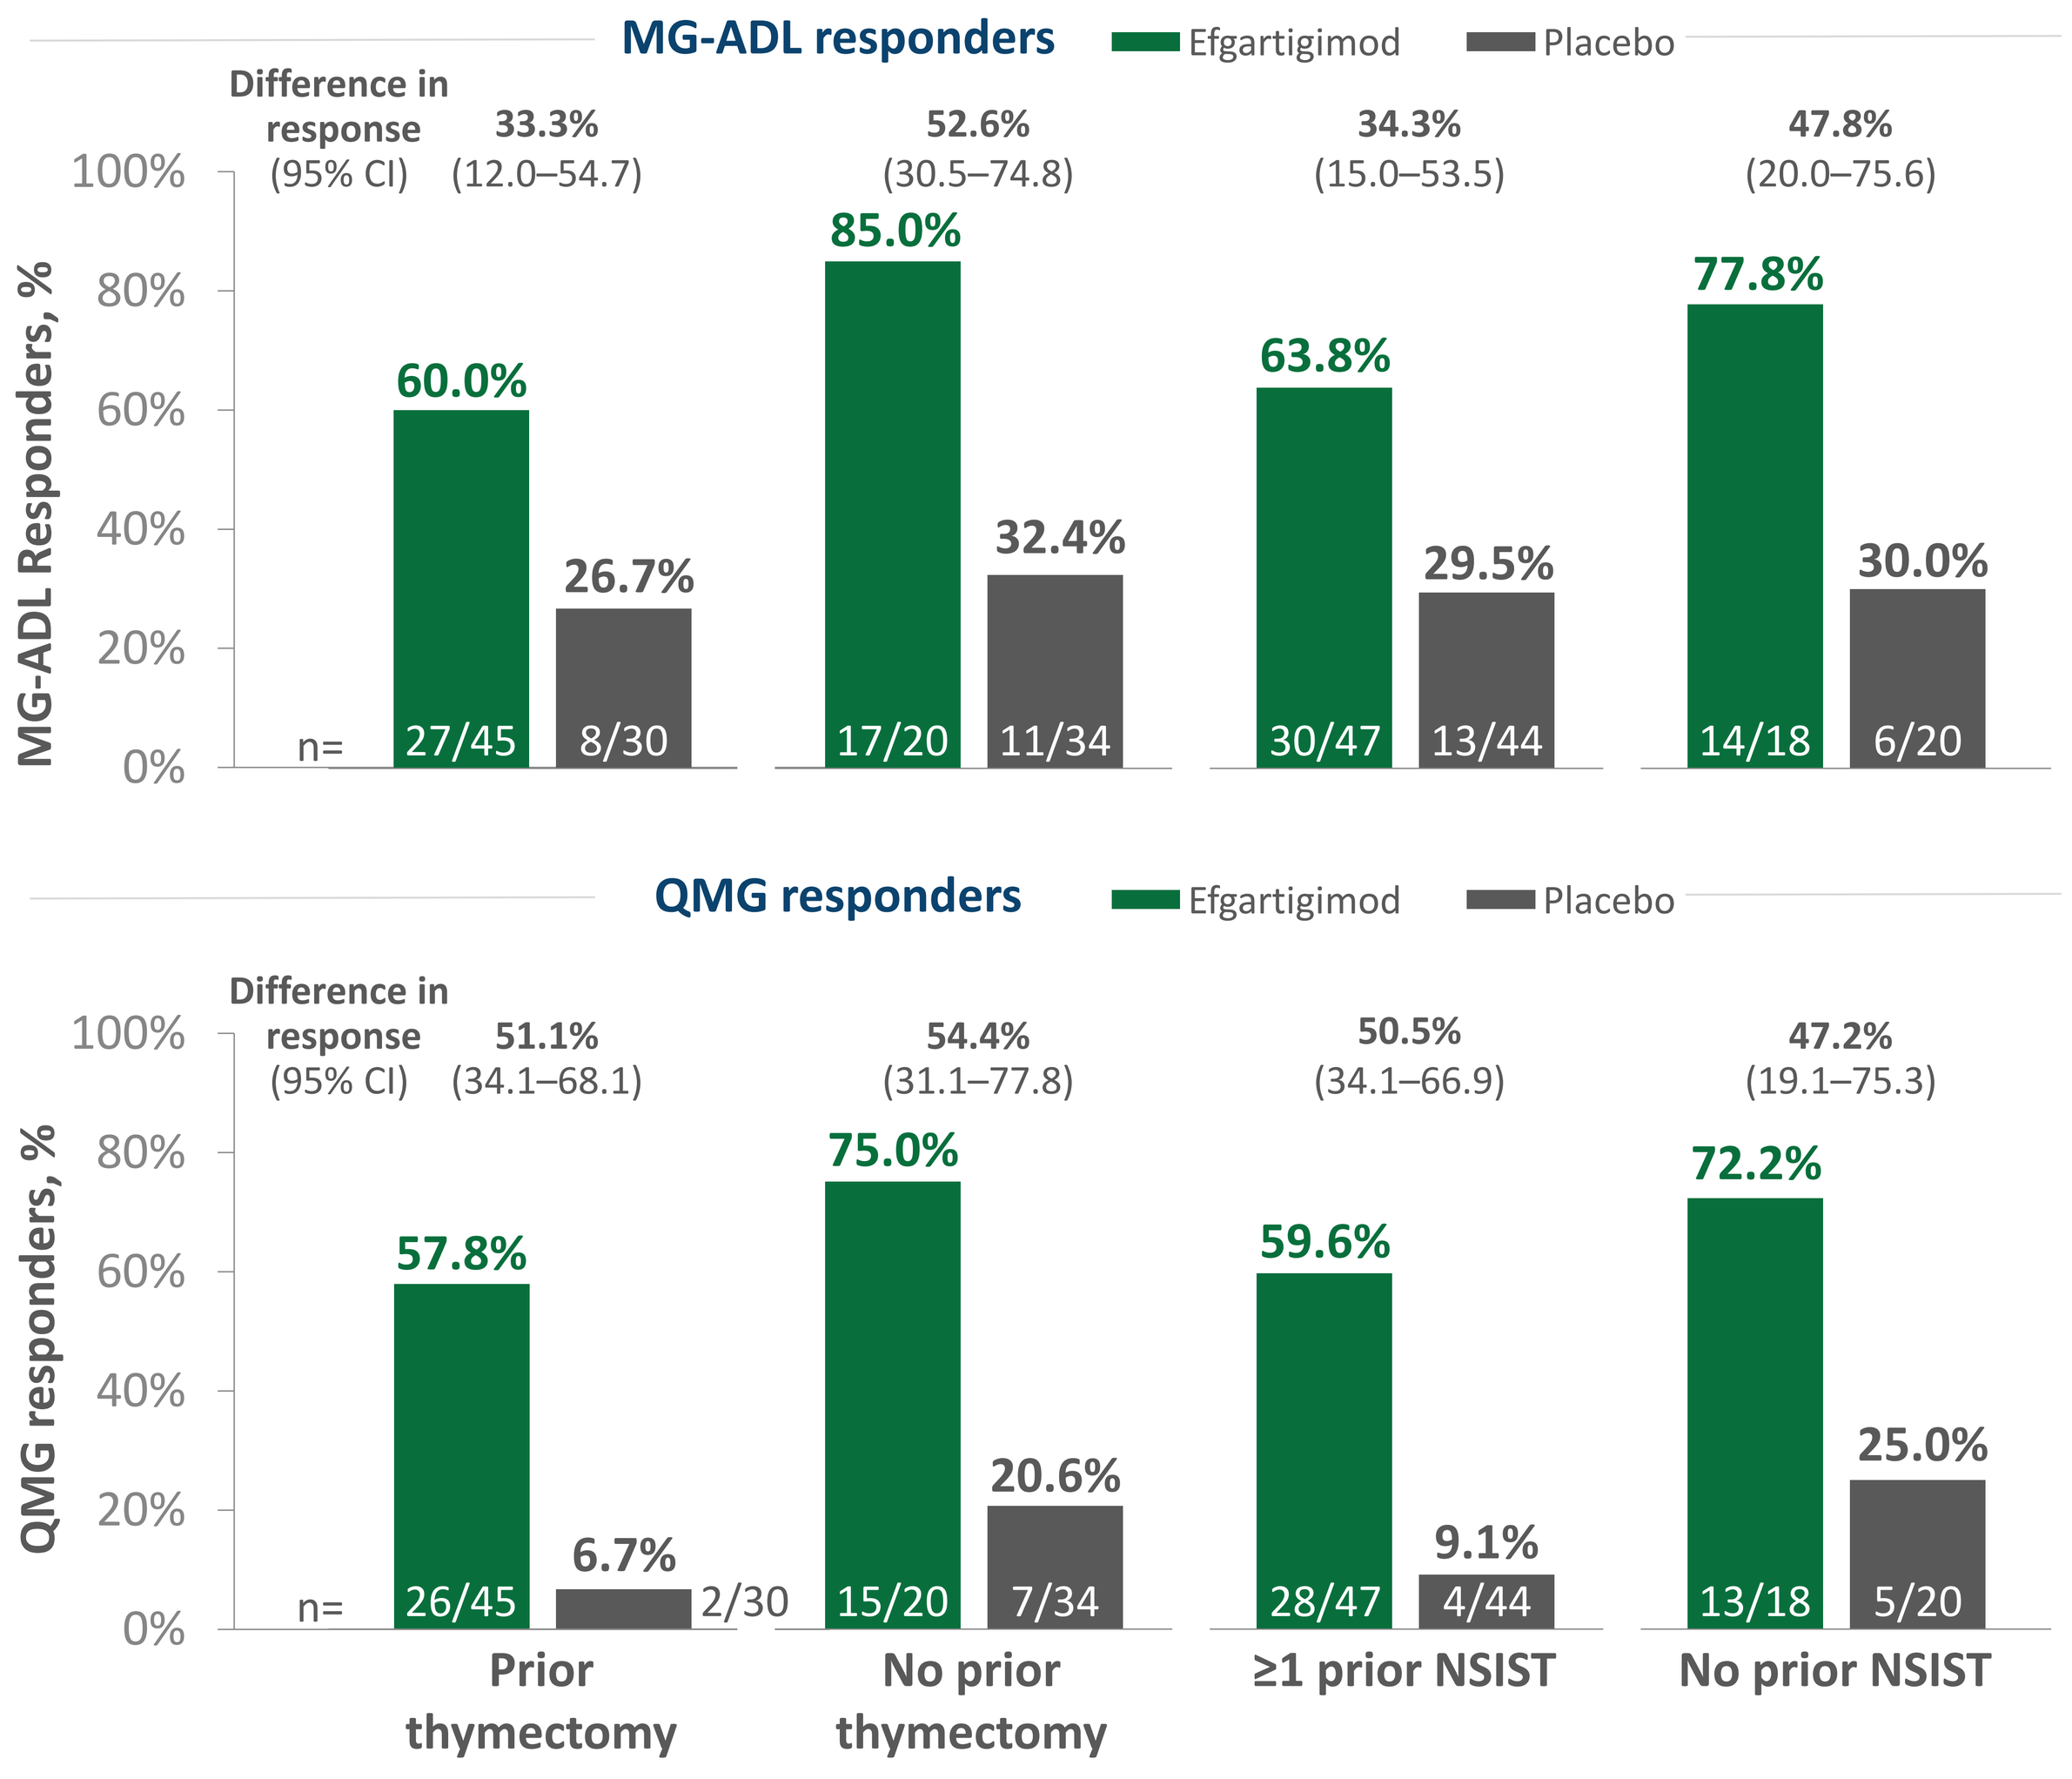

MG-ADL responders
Efgartigimod
Placebo
Difference in response (95% CI)
33.3% (12.0–54.7)
52.6% (30.5–74.8)
34.3% (15.0–53.5)
47.8% (20.0–75.6)
MG-ADL Responders, %
60.0%
26.7%
85.0%
32.4%
63.8%
29.5%
77.8%
30.0%
n= 27/45 8/30
17/20 11/34
30/47 13/44
14/18 6/20
QMG responders
51.1% (34.1–68.1)
54.4% (31.1–77.8)
50.5% (34.1–66.9)
47.2% (19.1–75.3)
QMG responders, %
57.8%
6.7%
75.0%
20.6%
59.6%
9.1%
72.2%
25.0%
n= 26/45 2/30
15/20 7/34
28/47 4/44
13/18 5/20
Prior thymectomy
No prior thymectomy
≥1 prior NSIST
No prior NSIST

Figure 4. Proportion of MG-ADL and QMG Responders Among Patients Without Prior Treatment Failures[a]
AChR-Ab+, Cycles 1 and 2

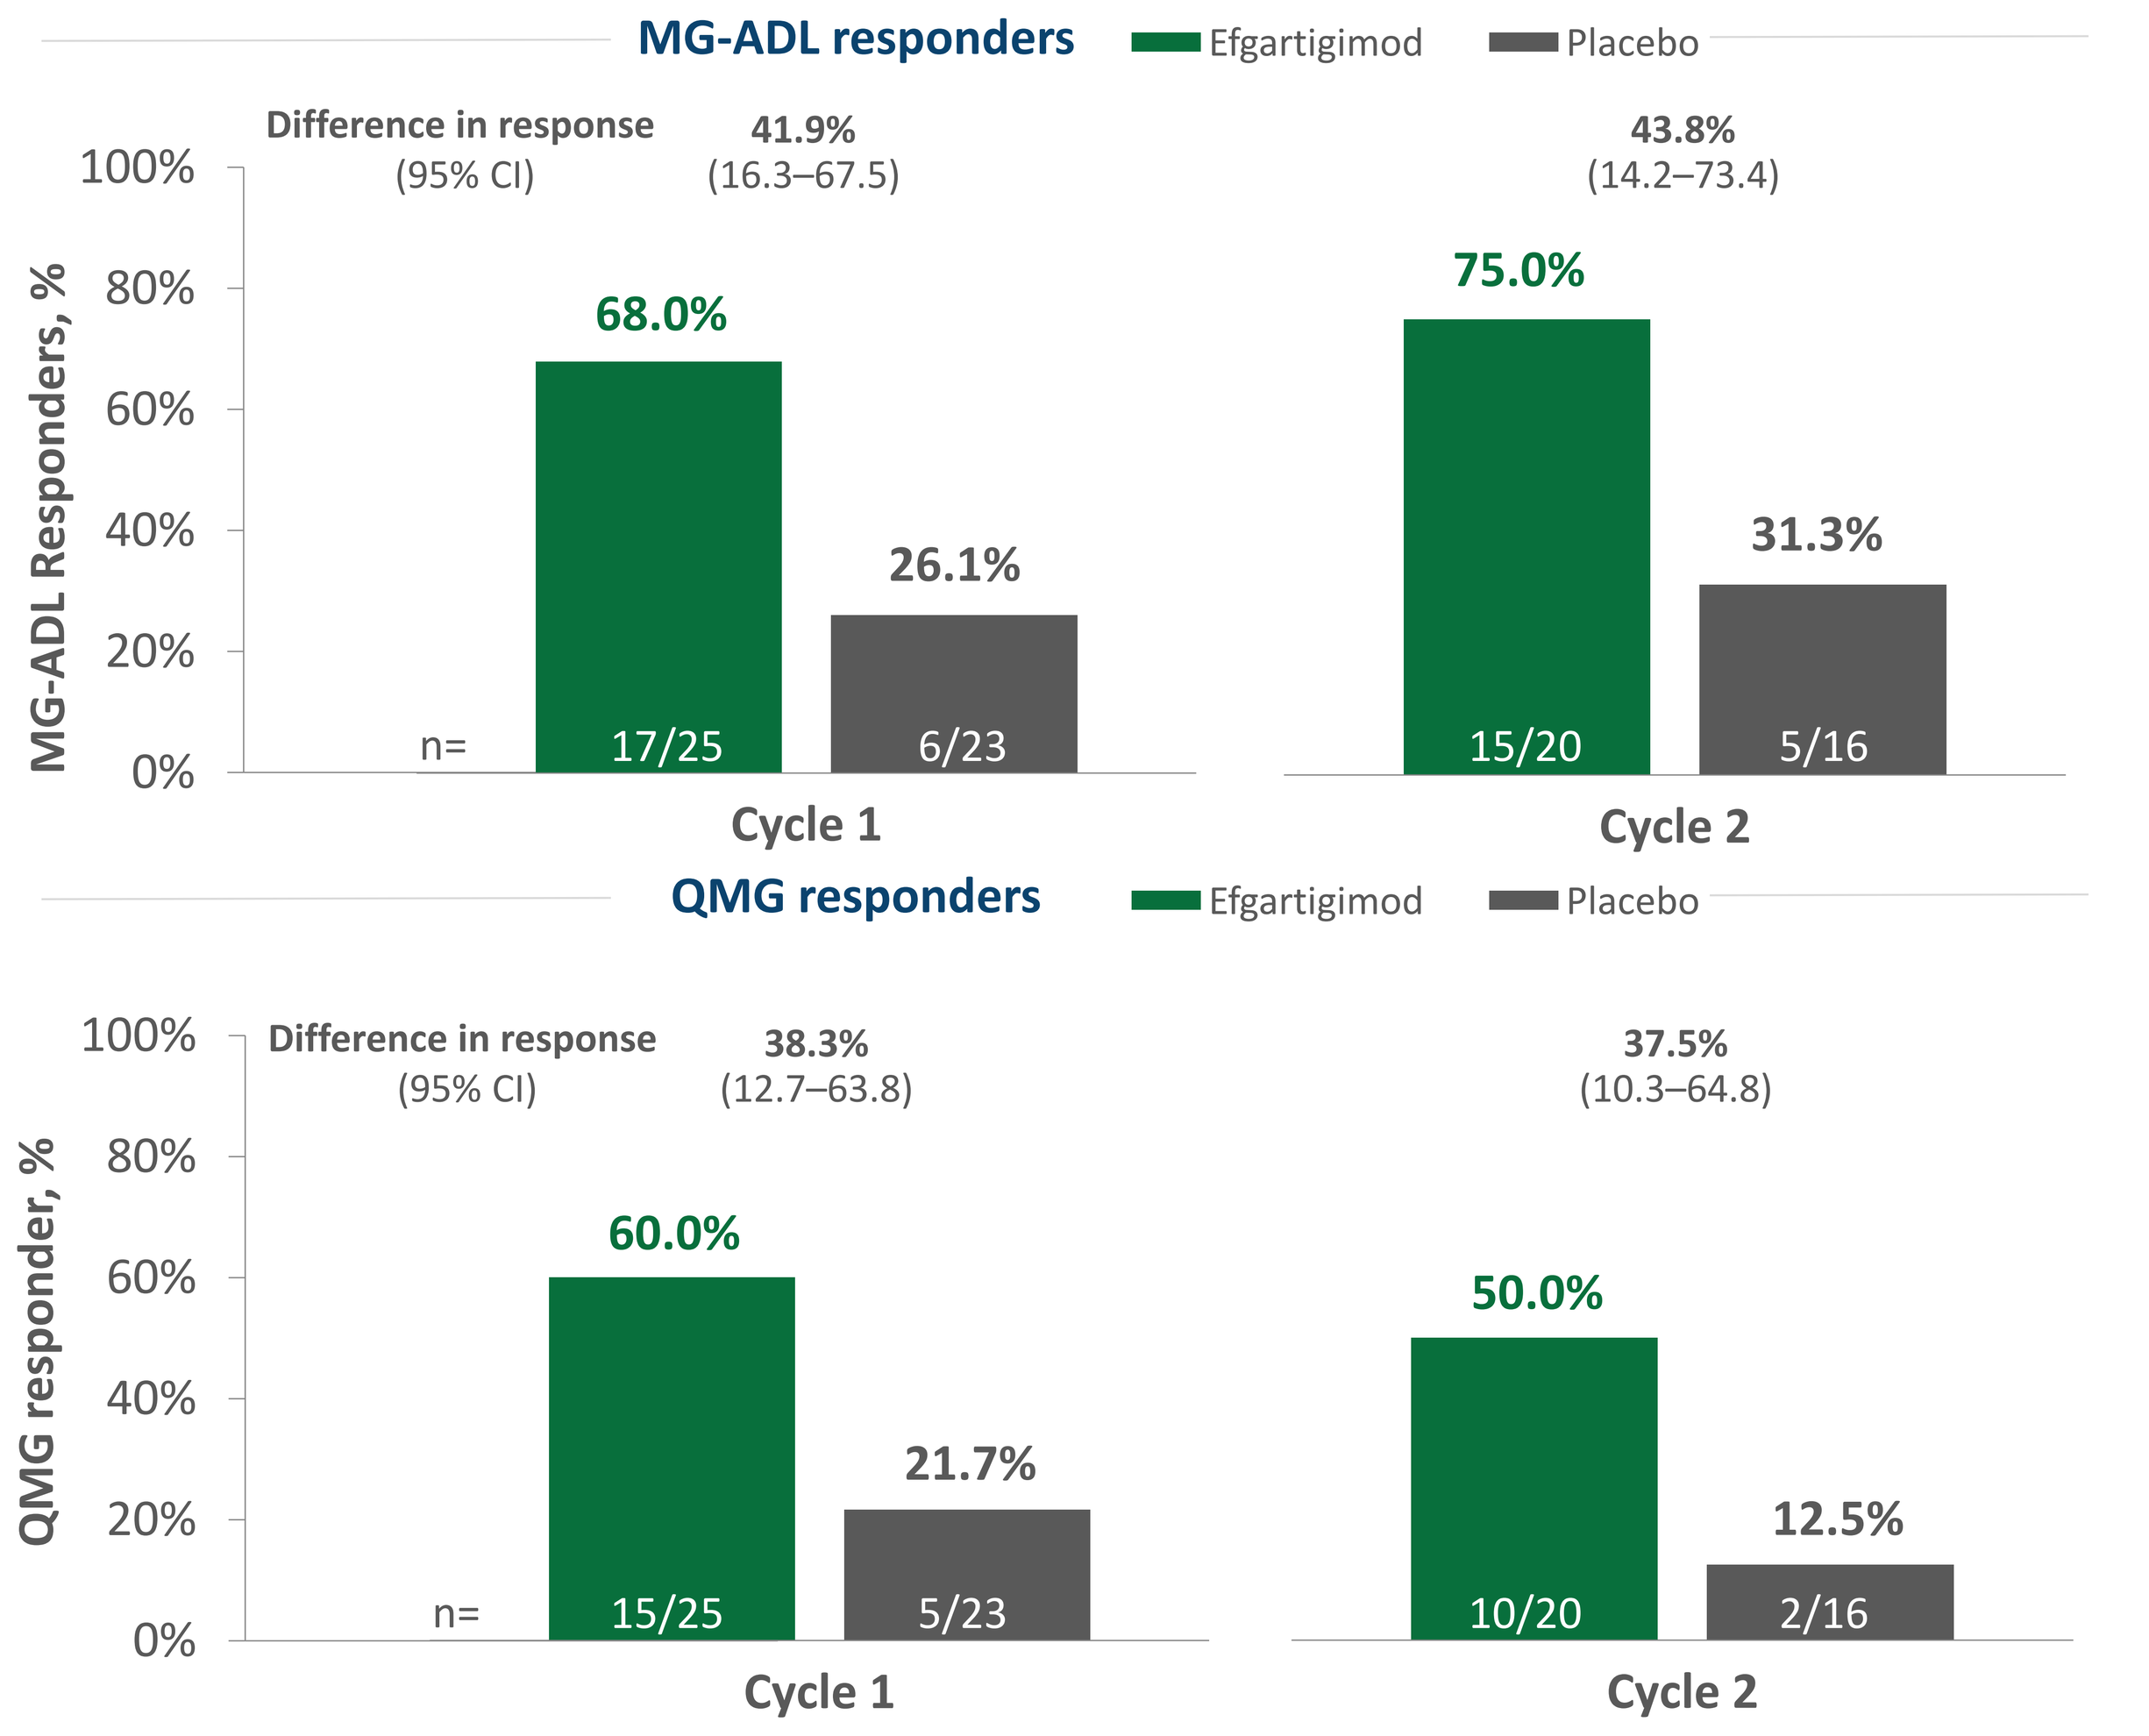

MG-ADL responders
Efgartigimod
Placebo
Difference in response (95% CI)
41.9% (16.3–67.5)
43.8% (14.2–73.4)
MG-ADL Responders, %
68.0%
26.1%
75.0%
31.3%
n= 17/25 6/23
15/20 5/16
Cycle 1
Cycle 2
QMG responders
38.3% (12.7–63.8)
37.5% (10.3–64.8)
QMG responder, %
60.0%
21.7%
50.0%
12.5%
n= 15/25 5/23
10/20 2/16

[a]Prior exposure to ≤2 immunosuppressive therapies, and not requiring PLEX or IVIg multiple times in the preceding year.

ABBREVIATIONS

AChEI, acetylcholinesterase inhibitor; AChR-Ab+, acetylcholine receptor antibody seropositive; AE, adverse event; FcRn, neonatal Fc receptor; Ig, immunoglobulin; IVIg, intravenous immunoglobulin; MG-ADL, Myasthenia Gravis Activities of Daily Living; NSIST, nonsteroidal immunosuppressive therapy; PLEX, plasma exchange; QMG, Quantitative Myasthenia Gravis; SAE, serious adverse event.

REFERENCES

1. Sesarman A, et al. *Cell Mol Life Sci*. 2010;67(15):2533-2550. **2.** Ulrichts P, et al. *J Clin Invest*. 2018;128(10):4372-4386. **3.** Vaccaro C, et al. *Nat Biotech*. 2005;23(10):1283-1288. **4.** Howard JF Jr, et al. *Lancet Neurol*. 2021;20(7):526-536. **5.** Nixon AE, et al. *Front Immunol.* 2015;6:176. **6.** Ward ES, et al. *Front Immunol.* 2022;13:892534.

ACKNOWLEDGMENTS AND DISCLOSURES: The authors gratefully acknowledge the ADAPT and ADAPT+ trial participants and investigators. **ER, EB,** and **RK** are employees of argenx BV; **VB** has received research support from CSL, Grifols SA, UCB, Bionevia, Shire, and Octapharma; **TV** has served as a speaker for Alexion Pharmaceuticals, Inc and argenx BV. He performed consulting work for Alexion Pharmaceuticals, argenx BV, and UCB and participated in trials in MG sponsored by National Institutes of Health, Alexion Pharmaceuticals, Inc, argenx BV, Ra Pharmaceuticals Inc/UCB, Viela Bio, Inc (now Horizon Therapeutics plc), Regeneron, Sanofi, Cartesian Therapeutics, and Grifols SA; **JFH** has received research support (paid to his institution) from Alexion Pharmaceuticals, Inc, argenx BV, Cartesian Therapeutics, the Centers for Disease Control and Prevention, Myasthenia Gravis Foundation of America, Muscular Dystrophy Association, National Institutes of Health (including the National Institute of Neurological Disorders and Stroke and the National Institute of Arthritis and Musculoskeletal and Skin Diseases), Patient-Centered Outcomes Research Institute, Ra Pharmaceuticals Inc (now UCB), and Takeda Pharmaceuticals; honoraria from Alexion Pharmaceuticals, Inc, argenx BV, Immunovant, Inc, Ra Pharmaceuticals Inc (now UCB), Regeneron Pharmaceuticals Inc, Sanofi US, and Viela Bio, Inc (now Horizon Therapeutics plc); and nonfinancial support from Alexion Pharmaceuticals, Inc, argenx BV, Ra Pharmaceuticals Inc (now UCB), and Toleranzia AB. Medical writing and editorial support for this presentation was provided by PRECISION Value and Health and funded by argenx BV.

argenx

Presented at the Muscular Dystrophy Association (MDA) Clinical & Scientific Conference; March 19–22, 2023; Dallas, TX, USA