314

An Open-Label Study to Assess the Clinical Efficacy and Safety of Additional Dosing Regimens of Efgartigimod IV: ADAPT-NXT, a Phase 3b Trial to Further Individualize Treatment Options for Patients With Generalized Myasthenia Gravis

Gregory Sahagian,[1] Yessar Hussain,[2] Marc H. Feinberg,[3] Ali A. Habib,[4] Thomas Skripuletz,[5] Tobias Ruck,[6] Edward Brauer,[7] Deborah Gelinas,[7] Li Liu,[7] Jana Podhorna,[7] Renato Mantegazza[8]

[1]The Neurology Center of Southern California, Carlsbad, California, USA; [2]Austin Neuromuscular Center, Austin, Texas, USA; [3]South Florida Neurology Associates, Boca Raton, Florida, USA; [4]Department of Neurology, University of California, Irvine, Irvine, California, USA; [5]Hannover Medical School, Hannover, Germany; [6]Department of Neurology, Medical Faculty, Heinrich-Heine-University, Düsseldorf, Germany; [7]argenx, Ghent, Belgium; [8]Department of Neuroimmunology and Neuromuscular Diseases, Fondazione IRCCS Istituto Neurologico Carlo Besta, Milan, Italy

Introduction

Efgartigimod Mechanism of Action: Blocking Neonatal Fc Receptor[1-4]

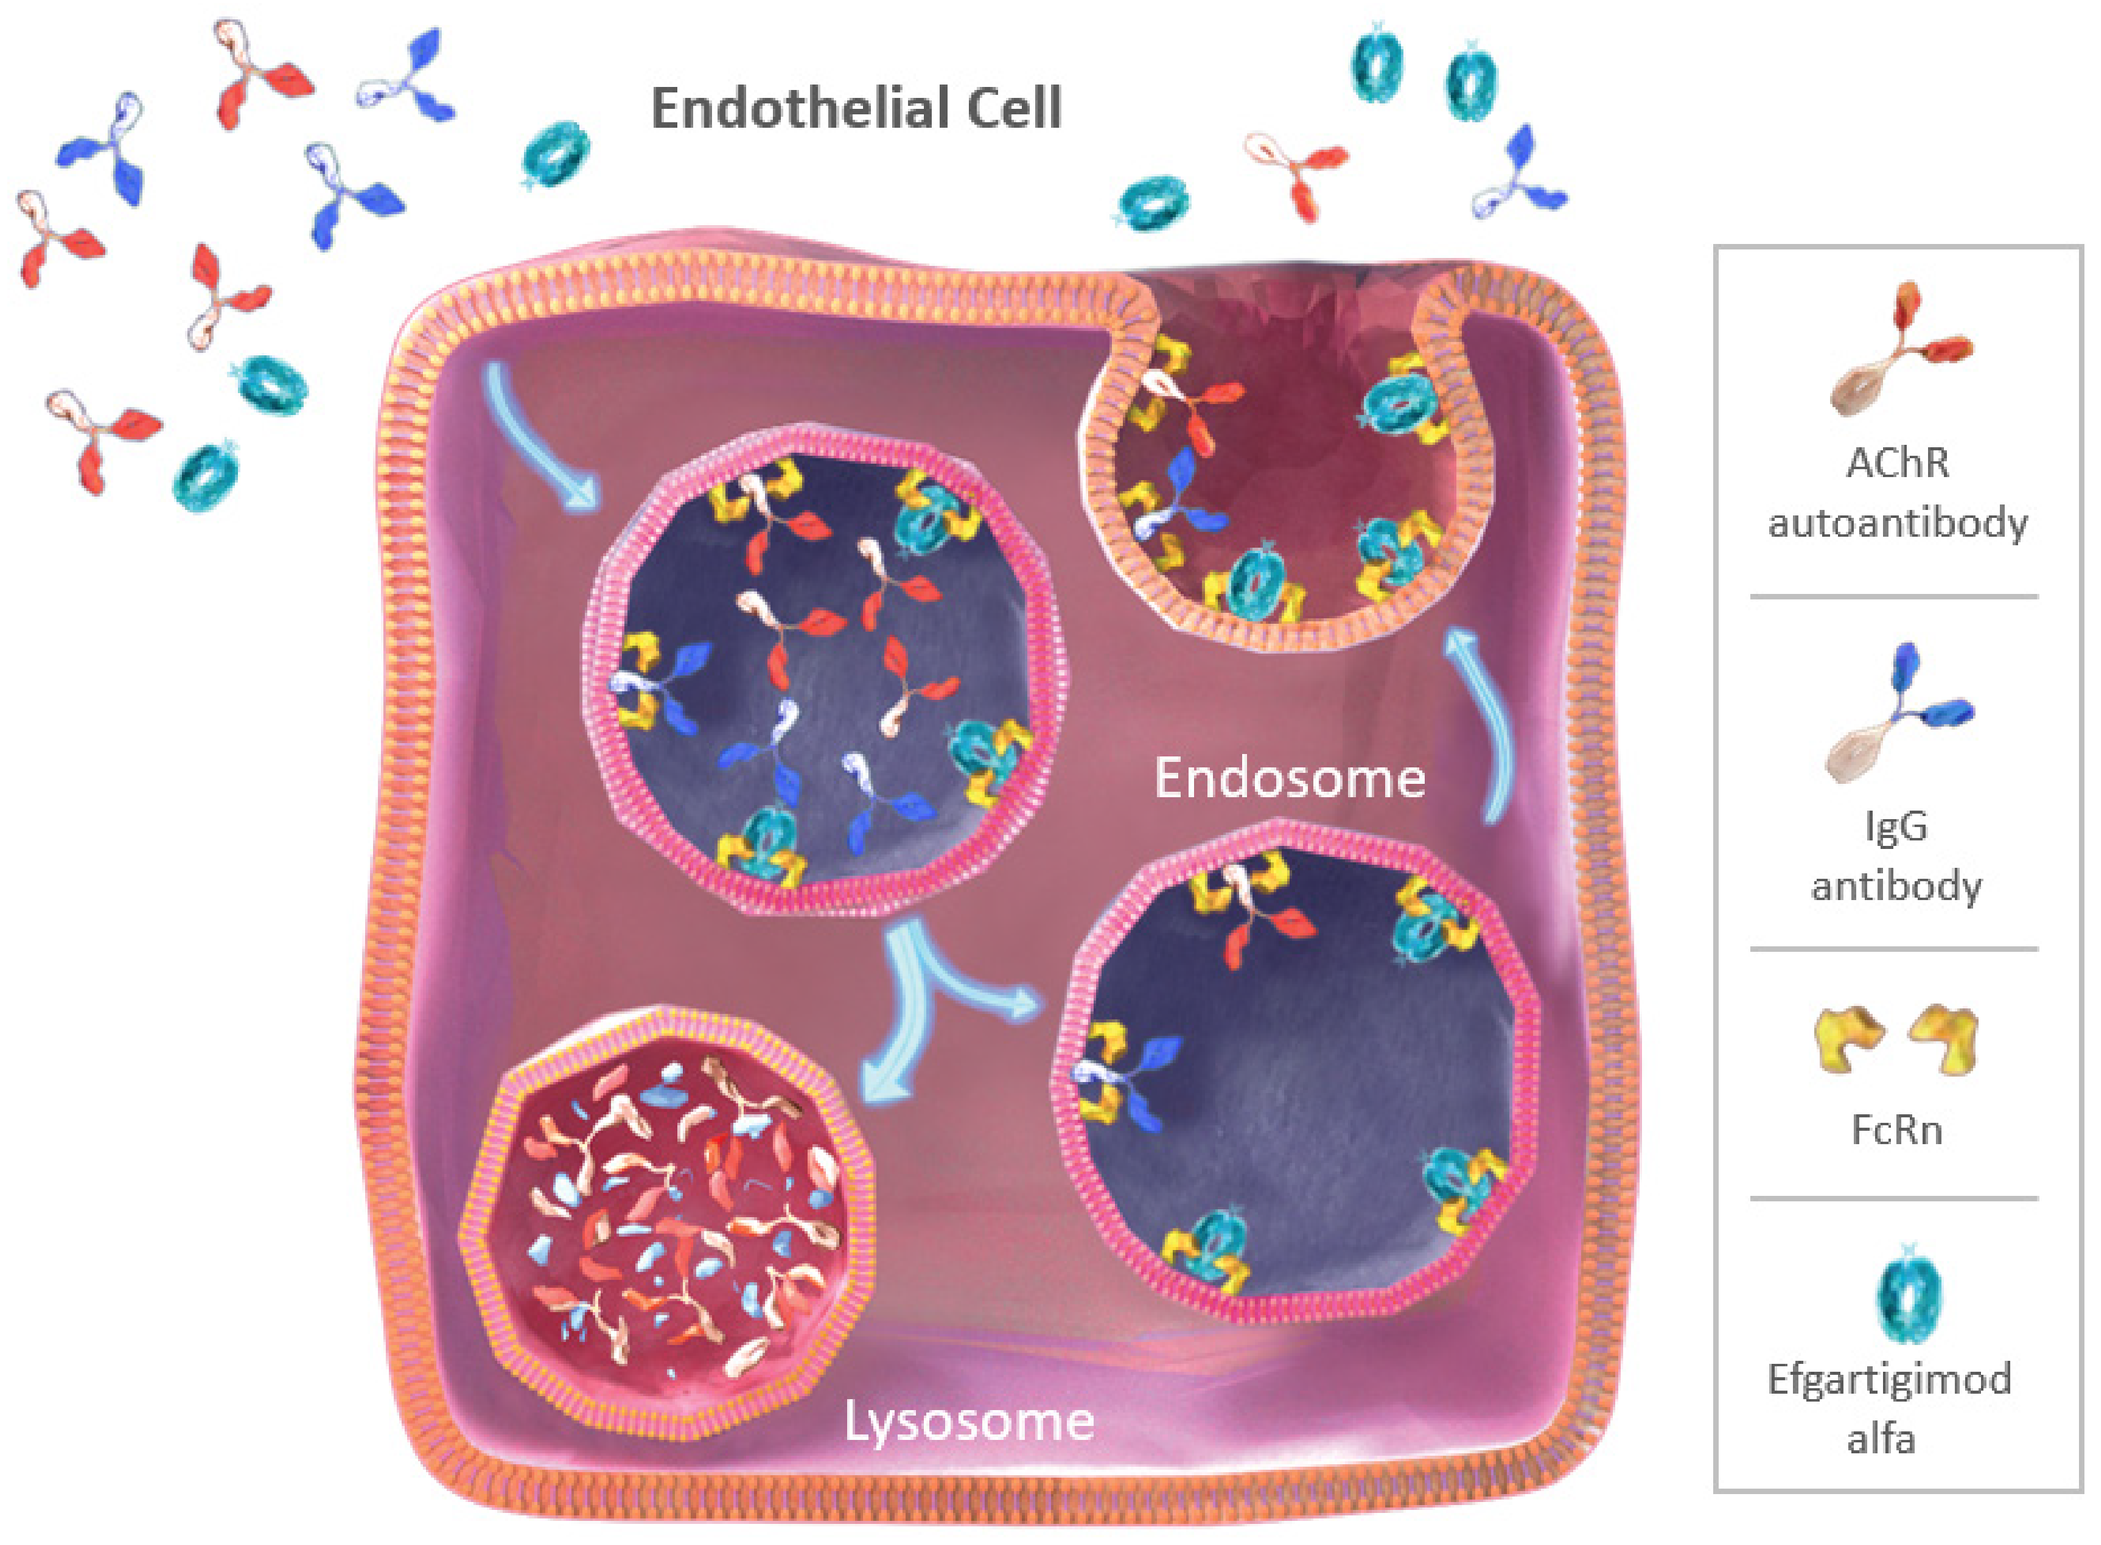

Endothelial Cell
Endosome
Lysosome
AChR autoantibody
IgG antibody
FcRn
Efgartigimod alfa

Image adapted from Kang TH, Jung ST. Boosting therapeutic potency of antibodies by taming Fc domain functions. *Exp Mol Med.* 2019;51:1–9 and distributed under the terms of the Creative Commons CC-BY license (https://creativecommons.org/licenses/by/4.0/)

- Neonatal Fc receptor (FcRn) recycles immunoglobulin G (IgG), extending its half-life and maintaining its serum concentration[1]
- Efgartigimod is a human IgG1 Fc fragment, a natural ligand of FcRn, engineered for increased affinity to FcRn[2]
- Efgartigimod was designed to outcompete endogenous IgG, preventing recycling and promoting lysosomal degradation of IgG[2,3]
 - Targeted reduction of all IgG subtypes
 - No impact on IgM or IgA
 - No reduction in albumin and no increase in cholesterol levels
 - No impact on IgG production or ability to mount an immune response

Generalized Myasthenia Gravis: An IgG-Mediated Autoimmune Disease[2]

- Generalized myasthenia gravis (gMG) is a rare, chronic, autoimmune disease mediated by pathogenic IgG autoantibodies that cause debilitating and potentially life-threatening muscle weakness
- IgG autoantibodies against acetylcholine receptor (AChR) have been identified in 85% of adults with gMG
- IgG autoantibodies are directly pathogenic in gMG, causing failure of neuromuscular transmission by targeting receptors and proteins of the neuromuscular junction

Efgartigimod Has Been Shown to Effectively Reduce IgG Antibodies in Adult Clinical Studies[2,5]

- Individualized cyclic administration of efgartigimod demonstrated safety and efficacy in randomized, double-blinded, placebo-controlled trials (phase 2, NCT02965573, N=24; phase 3, NCT03669588, N=167) in patients with gMG[2,5]
- The phase 3 trial (ADAPT) demonstrated efficacy, with the efgartigimod intravenous (IV) treatment arm reporting a significantly higher percentage of Myasthenia Gravis Activities of Daily Living (MG-ADL) responders (68%) compared with the placebo arm (37%) ($P<.0001$) in cycle 1, regardless of antibody status. Similar responses were seen in Quantitative Myasthenia Gravis responders[2]
- Efgartigimod 10 mg/kg administered as 4 infusions per cycle (1 infusion per week), with a minimum of 7 weeks from the first infusion of the previous cycle to the first infusion of the subsequent cycle (if given), demonstrated a favorable benefit-risk ratio in both the phase 2 and the phase 3 trials. The majority of reported treatment-emergent adverse events were mild or moderate in severity[2,5]

Objective

- To evaluate the efficacy and safety of efgartigimod 10 mg/kg IV administered in different dosing regimens to help physicians further individualize treatment for patients with gMG

Key Eligibility Criteria

Inclusion Criteria

- ≥18 years of age
- AChR antibody–seropositive patients with gMG
- MG-ADL total score ≥5, with >50% of the score due to nonocular symptoms
- Concomitant gMG therapy permitted, but not required (nonsteroidal immunosuppressive drugs [NSIDs], steroids, and/or acetylcholinesterase inhibitors; stable dose for ≥1 month before screening for NSIDs and/or corticosteroids)

Exclusion Criteria

- Total IgG <6 g/L at screening
- Use of IV or subcutaneous Ig within 14 days, eculizumab within 1 month, any other investigational product within 3 months or 5 half-lives, monoclonal antibody within 5 half-lives, or rituximab within 6 months of screening
- Received a live or live-attenuated vaccine <28 days before screening
- History of malignancy
- Received a thymectomy <3 months before screening
- Active infection

Design

ADAPT NXT (ARGX-113-2003) Trial Design

Efgartigimod IV phase 3b, multicenter, randomized, open-label, parallel-group trial in patients with gMG

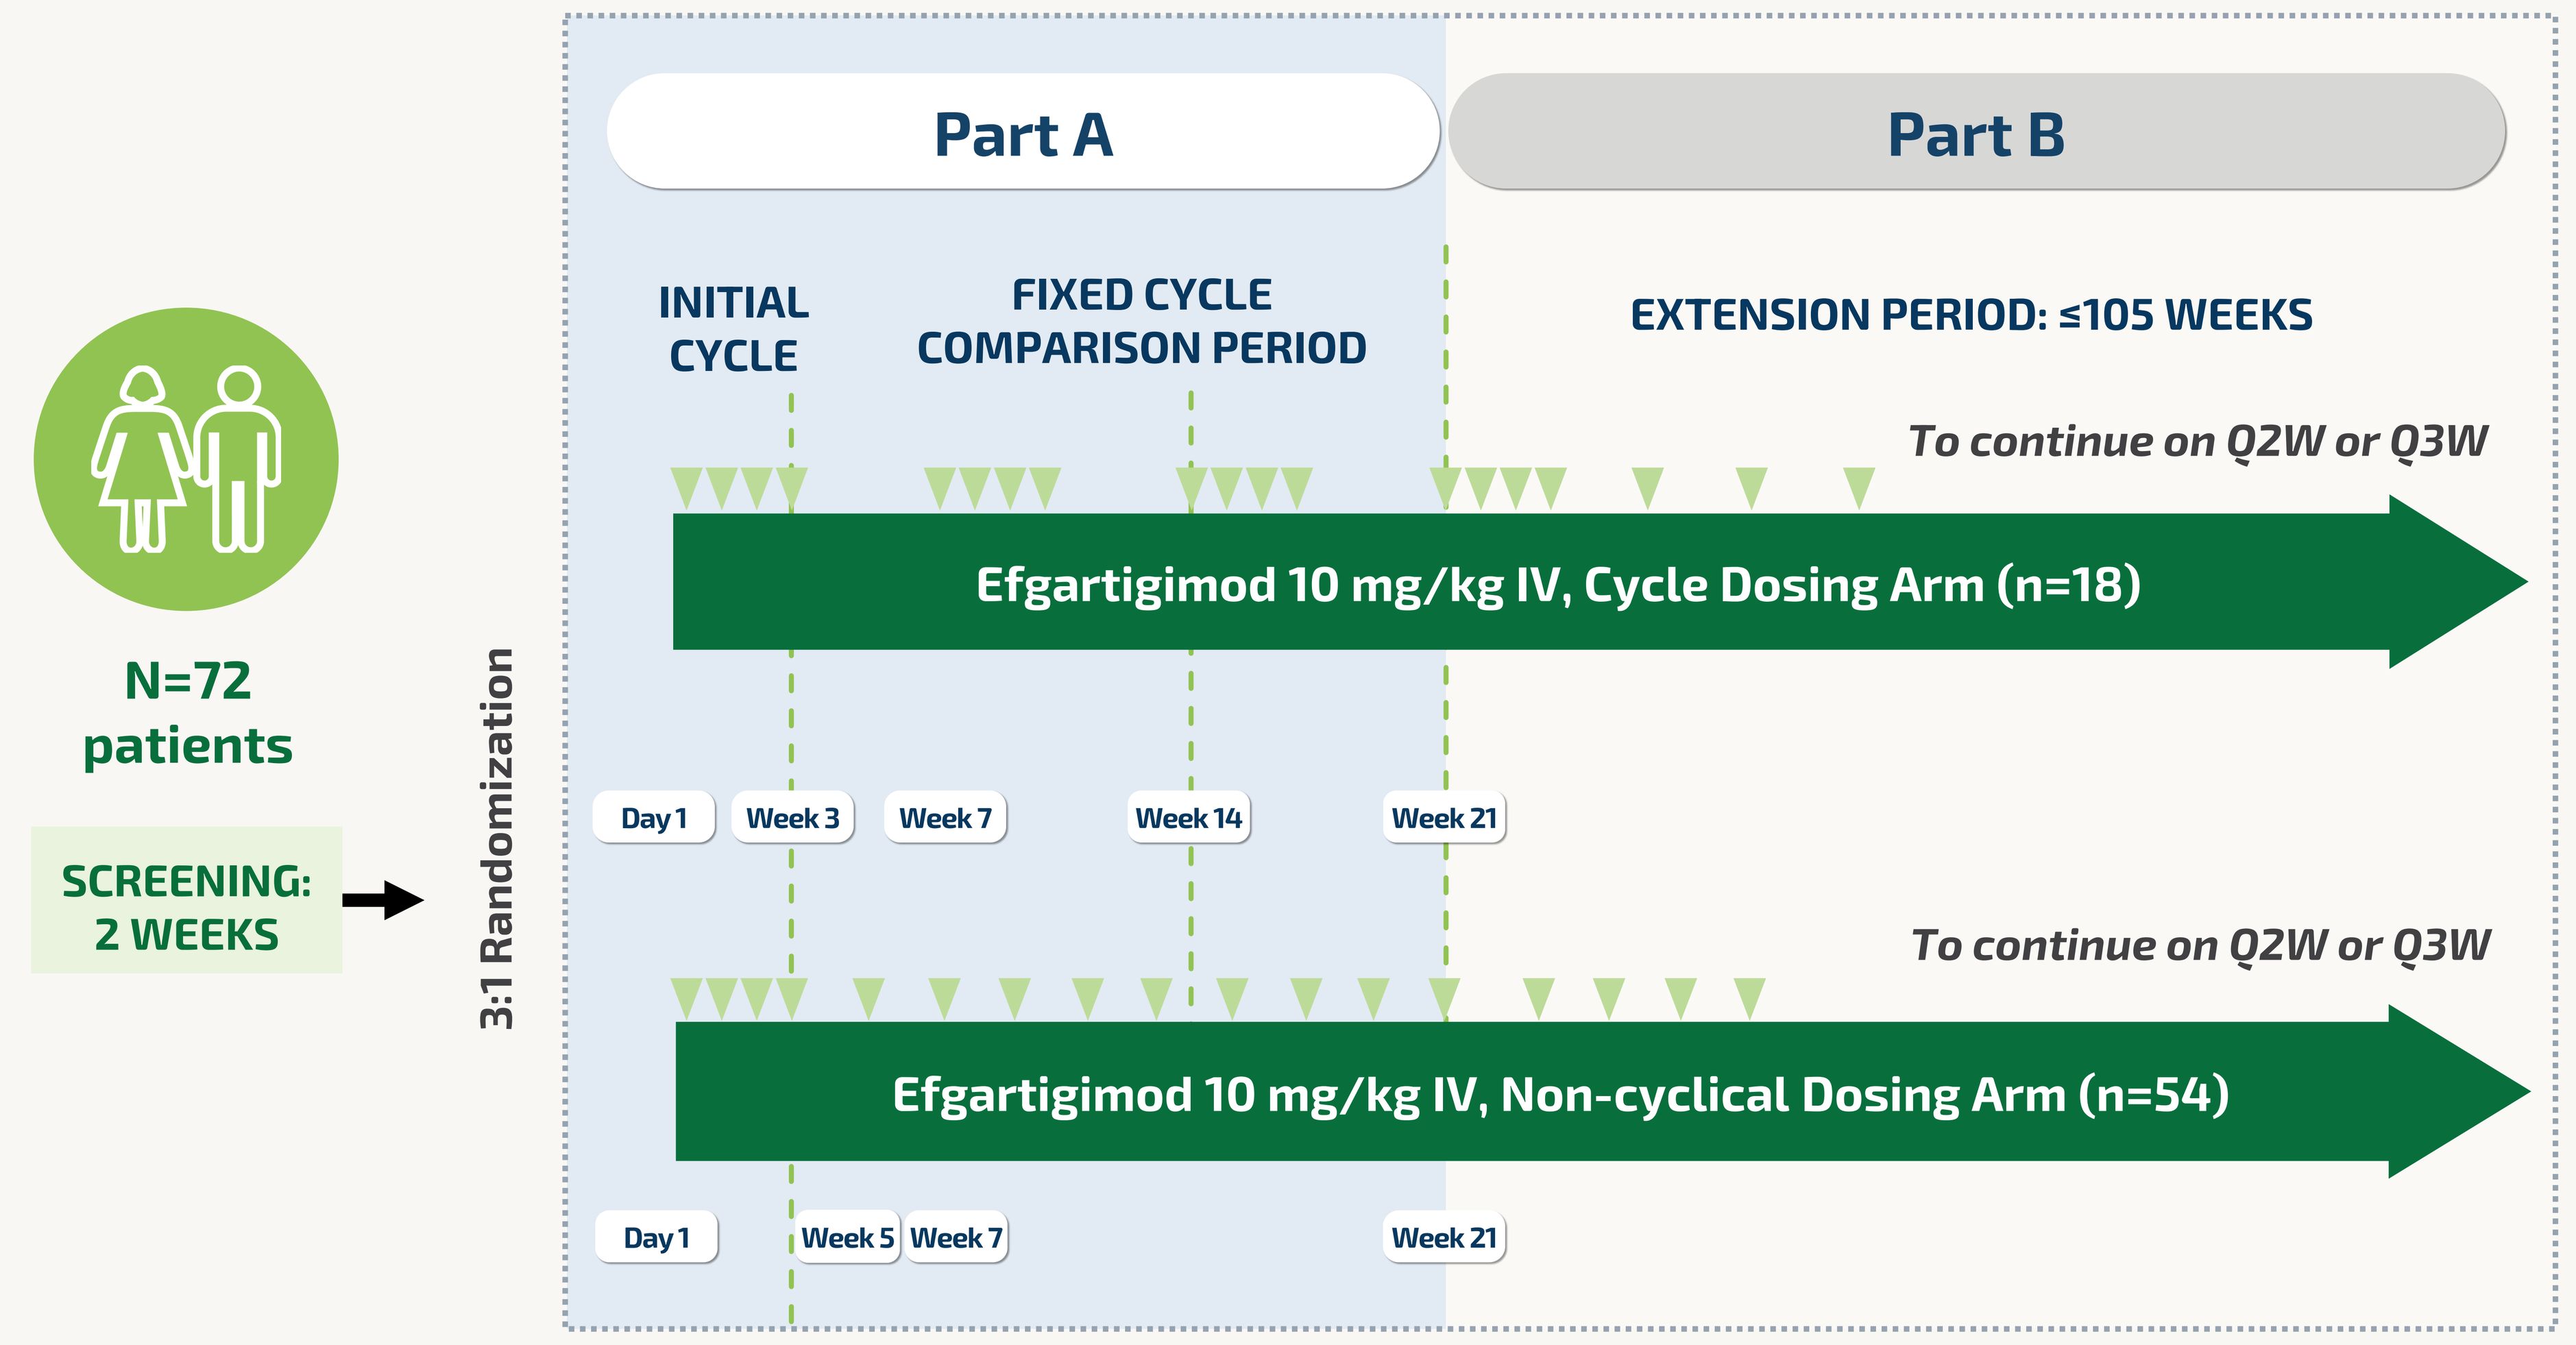

N=72 patients
SCREENING: 2 WEEKS
3:1 Randomization
Part A
Part B
INITIAL CYCLE
FIXED CYCLE COMPARISON PERIOD
EXTENSION PERIOD: ≤105 WEEKS
To continue on Q2W or Q3W
Efgartigimod 10 mg/kg IV, Cycle Dosing Arm (n=18)
Day 1
Week 3
Week 7
Week 14
Week 21
To continue on Q2W or Q3W
Efgartigimod 10 mg/kg IV, Non-cyclical Dosing Arm (n=54)
Day 1
Week 5
Week 7
Week 21

gMG, generalized myasthenia gravis; IV, intravenous; Q2W, every 2 weeks; Q3W, every 3 weeks.
Trial ARGX-113-2003 (ADAPT NXT) Clinical Trial Protocol v1.0, July 6, 2021.

Primary Endpoint

- Mean of the average MG-ADL total score change from Week 1 through Week 21 by regimen arm

Secondary Endpoints

- Incidence and severity of adverse events, serious adverse events, adverse events of special interest, laboratory test results, vital signs, and electrocardiogram results
- Change from baseline in the MG-ADL total score over time
- Percentage of participants who have a ≥2-, 3-, 4-, or 5-point improvement in MG-ADL total score over time
- Participants achieving minimal symptom expression, defined as MG-ADL total score 0–1, over time

Exploratory Endpoints

- Patient treatment satisfaction (TSQM-9 domain scores)
- Pharmacokinetic and pharmacodynamic effects
- Impact on quality of life (MG-QoL15r, EQ-5D-5L VAS, and Neuro-QoL)
- Feasibility of remote MG-ADL scale administration and receiving efgartigimod infusions off site

EQ-5D-5L VAS, EuroQoL 5-Dimension 5-Level visual analog scale; MG-QoL15r, Myasthenia Gravis Quality of Life 15-Item Questionnaire, Revised; Neuro-QoL, Adult Quality of Life in Neurological Disorders; TSQM-9, Treatment Satisfaction Questionnaire for Medication—9 Items.

Summary

Phase 3b, multicenter, randomized, open-label, parallel-group trial evaluating the efficacy and safety of different dosing regimens of efgartigimod IV in patients with gMG

ADAPT NXT will determine the efficacy, safety, and tolerability of efgartigimod 10 mg/kg IV administered in a non-cyclical dosing regimen

Recruitment is ongoing

Target: 72 patients (3:1 randomization ratio)

REFERENCES: **1.** Sesarman A, et al. *Cell Mol Life Sci.* 2010;67:2533–50. **2.** Howard JF Jr, et al. *Lancet Neurol.* 2021;20:526–36. **3.** Goebeler M, et al. *Br J Dermatol.* 2021;1–11. **4.** Wolfe GI, et al. *J Neurol Sci.* 2021;430:118074. **5.** Howard JF Jr, et al. *Neurology.* 2019;92:e2661–73.

DISCLOSURES: **GS:** Consulting fees/honoraria or support for meeting participation: Alexion Pharmaceuticals, Inc, argenx BV, UCB, Immunovant, Inc., Biogen Inc. **YH:** No disclosures to report. **MHF:** Consulting fees/speaker fees: argenx, abbvie, Allergan, Eisai, Catalyst, CAla Health. **AAH:** Research support: argenx, Alexion Pharmaceuticals, Inc, VielaBio, UCB Pharma, Genentech, Regeneron, Sanofi; Consulting fees: argenx, Alexion Pharmaceuticals, Inc, UCB. **TS:** Research support: Bristol-Myers Squibb, Sanofi; Consulting fees/honoraria: Alexion Pharmaceuticals, Inc, Alnylam, Bayer Vital, Biogen, Celgene, CSL Behring, Euroimmun, Janssen, Merck Serono, Novartis, Roche, Sanofi Aventis, Siemens. **TR:** Consulting fees/honoraria or support for meeting participation: Abbott, Alexion Pharmaceuticals, Inc, argenx BV, Biogen Inc., Celgene, Merck, Novartis, Roche, Teva, UCB. **EB**, **DG**, **LL**, **JP:** Employees of argenx. **RM:** Consulting fees/honoraria or support for meeting participation: Alexion Pharmaceuticals, Inc, argenx BV, Ra Pharmaceuticals Inc, BioMarin, Catalyst Pharmaceuticals, Inc, UCB, Teva, Merck, Roche, Biogen Inc.

FUNDING AND ACKNOWLEDGMENTS: The ADAPT-NXT study is funded by argenx. Formatting and editing assistance were provided by Alligent Europe (Envision Pharma Group), funded by argenx. Efgartigimod is an investigational agent being developed by argenx and has not been approved by the European Medicines Agency.

We gratefully acknowledge the clinicians, patient organizations, and scientists who have collaborated on the design of this trial

Presented at the 7th International Myology Congress; September 12–15, 2022; Nice, France