971

Clinical Outcomes, Disease Course, and QoL in Patients With Multifocal Motor Neuropathy: iMMersioN, Study in Progress

Stojan Perić,[1] Luis Querol,[2,3] Sadiq Altamimi,[4] Jamie Wood,[5] Inge Van de Walle,[5] Emma Persson,[5] Iris Van Hoomissen,[5] Gabriela Szmyd,[5] Miodrag Vujcic,[5] Marqus Hamwright,[5] Olivier Van de Steen,[5] Clémence Arvin-Bérod,[5] Jeffrey A. Allen[6]

[1]University of Belgrade, Faculty of Medicine, Neurology Clinic, University Clinical Center of Serbia, Belgrade, Serbia; [2]Department of Neurology, Neuromuscular Diseases Unit, Hospital de La Santa Creu I Sant Pau, Universitat Autònoma de Barcelona, Barcelona, Spain; [3]Centro de Investigación Biomédica en Red en Enfermedades Raras (CIBERER), Madrid, Spain; [4]The Neurology Group, Pomona, CA, USA; [5]argenx, Ghent, Belgium; [6]University of Minnesota, Department of Neurology, Minneapolis, MN, USA

INTRODUCTION

Multifocal Motor Neuropathy (MMN)

- MMN is a rare, peripheral, immune-mediated, chronic neuropathy, associated with axonal degeneration and progressive, disabling asymmetric limb weakness without sensory loss[1–3]
- MMN is driven by motor nerve conduction block due to IgM autoantibody-mediated complement activation[1–3]
- Patients with MMN typically have a normal life expectancy; however, ≤20% of patients experience relatively severe disability, predominantly in the upper limbs[3]
- Patients living with MMN report broad impacts on their daily lives, work, social life, and overall well-being[4]

iMMersioN Study Rationale

- Due to the **low prevalence of the disease** (at least 0.6 per 100,000 individuals),[1] observational data on patient experience and management of MMN in clinical practice are usually limited to small cohorts and retrospective analyses
- There is an opportunity to **further understand MMN** diagnosis, disease course and management, and to characterize the healthcare resource use of patients
- **iMMersioN** is a global, prospective, longitudinal study that will follow participants with MMN over time and collect data on clinical outcomes, HRQoL, and use of healthcare resources

STUDY DESIGN

iMMersioN (NCT05988073): A Multicenter, Prospective, Longitudinal Study in Adult Participants With MMN

Study population: ~200 participants with MMN receiving standard of care treatment

OBSERVATIONAL PHASE

Screening/ baseline → **Week 1–2** → **Month 3** → **Month 6** → **Every 3 months** → **Month 24**

UP TO 24 MONTHS

Visits take place approximately every 3 months, coinciding with regular MMN treatment visits; no investigational product will be administered

KEY ELIGIBILITY CRITERIA

Inclusion Criteria

- At least local legal age of consent for clinical studies when signing the informed consent form
- Capable of providing informed consent to participate in the study and complying with protocol requirements
- Diagnosis of MMN by a neuromuscular specialist or neurologist

Exclusion Criteria

- Participation in any clinical trial with an investigational medicinal product
- Presence of other medical condition that could affect the assessment of MMN

ENROLLMENT STATUS

- iMMersioN will be running in >115 sites in 20 countries

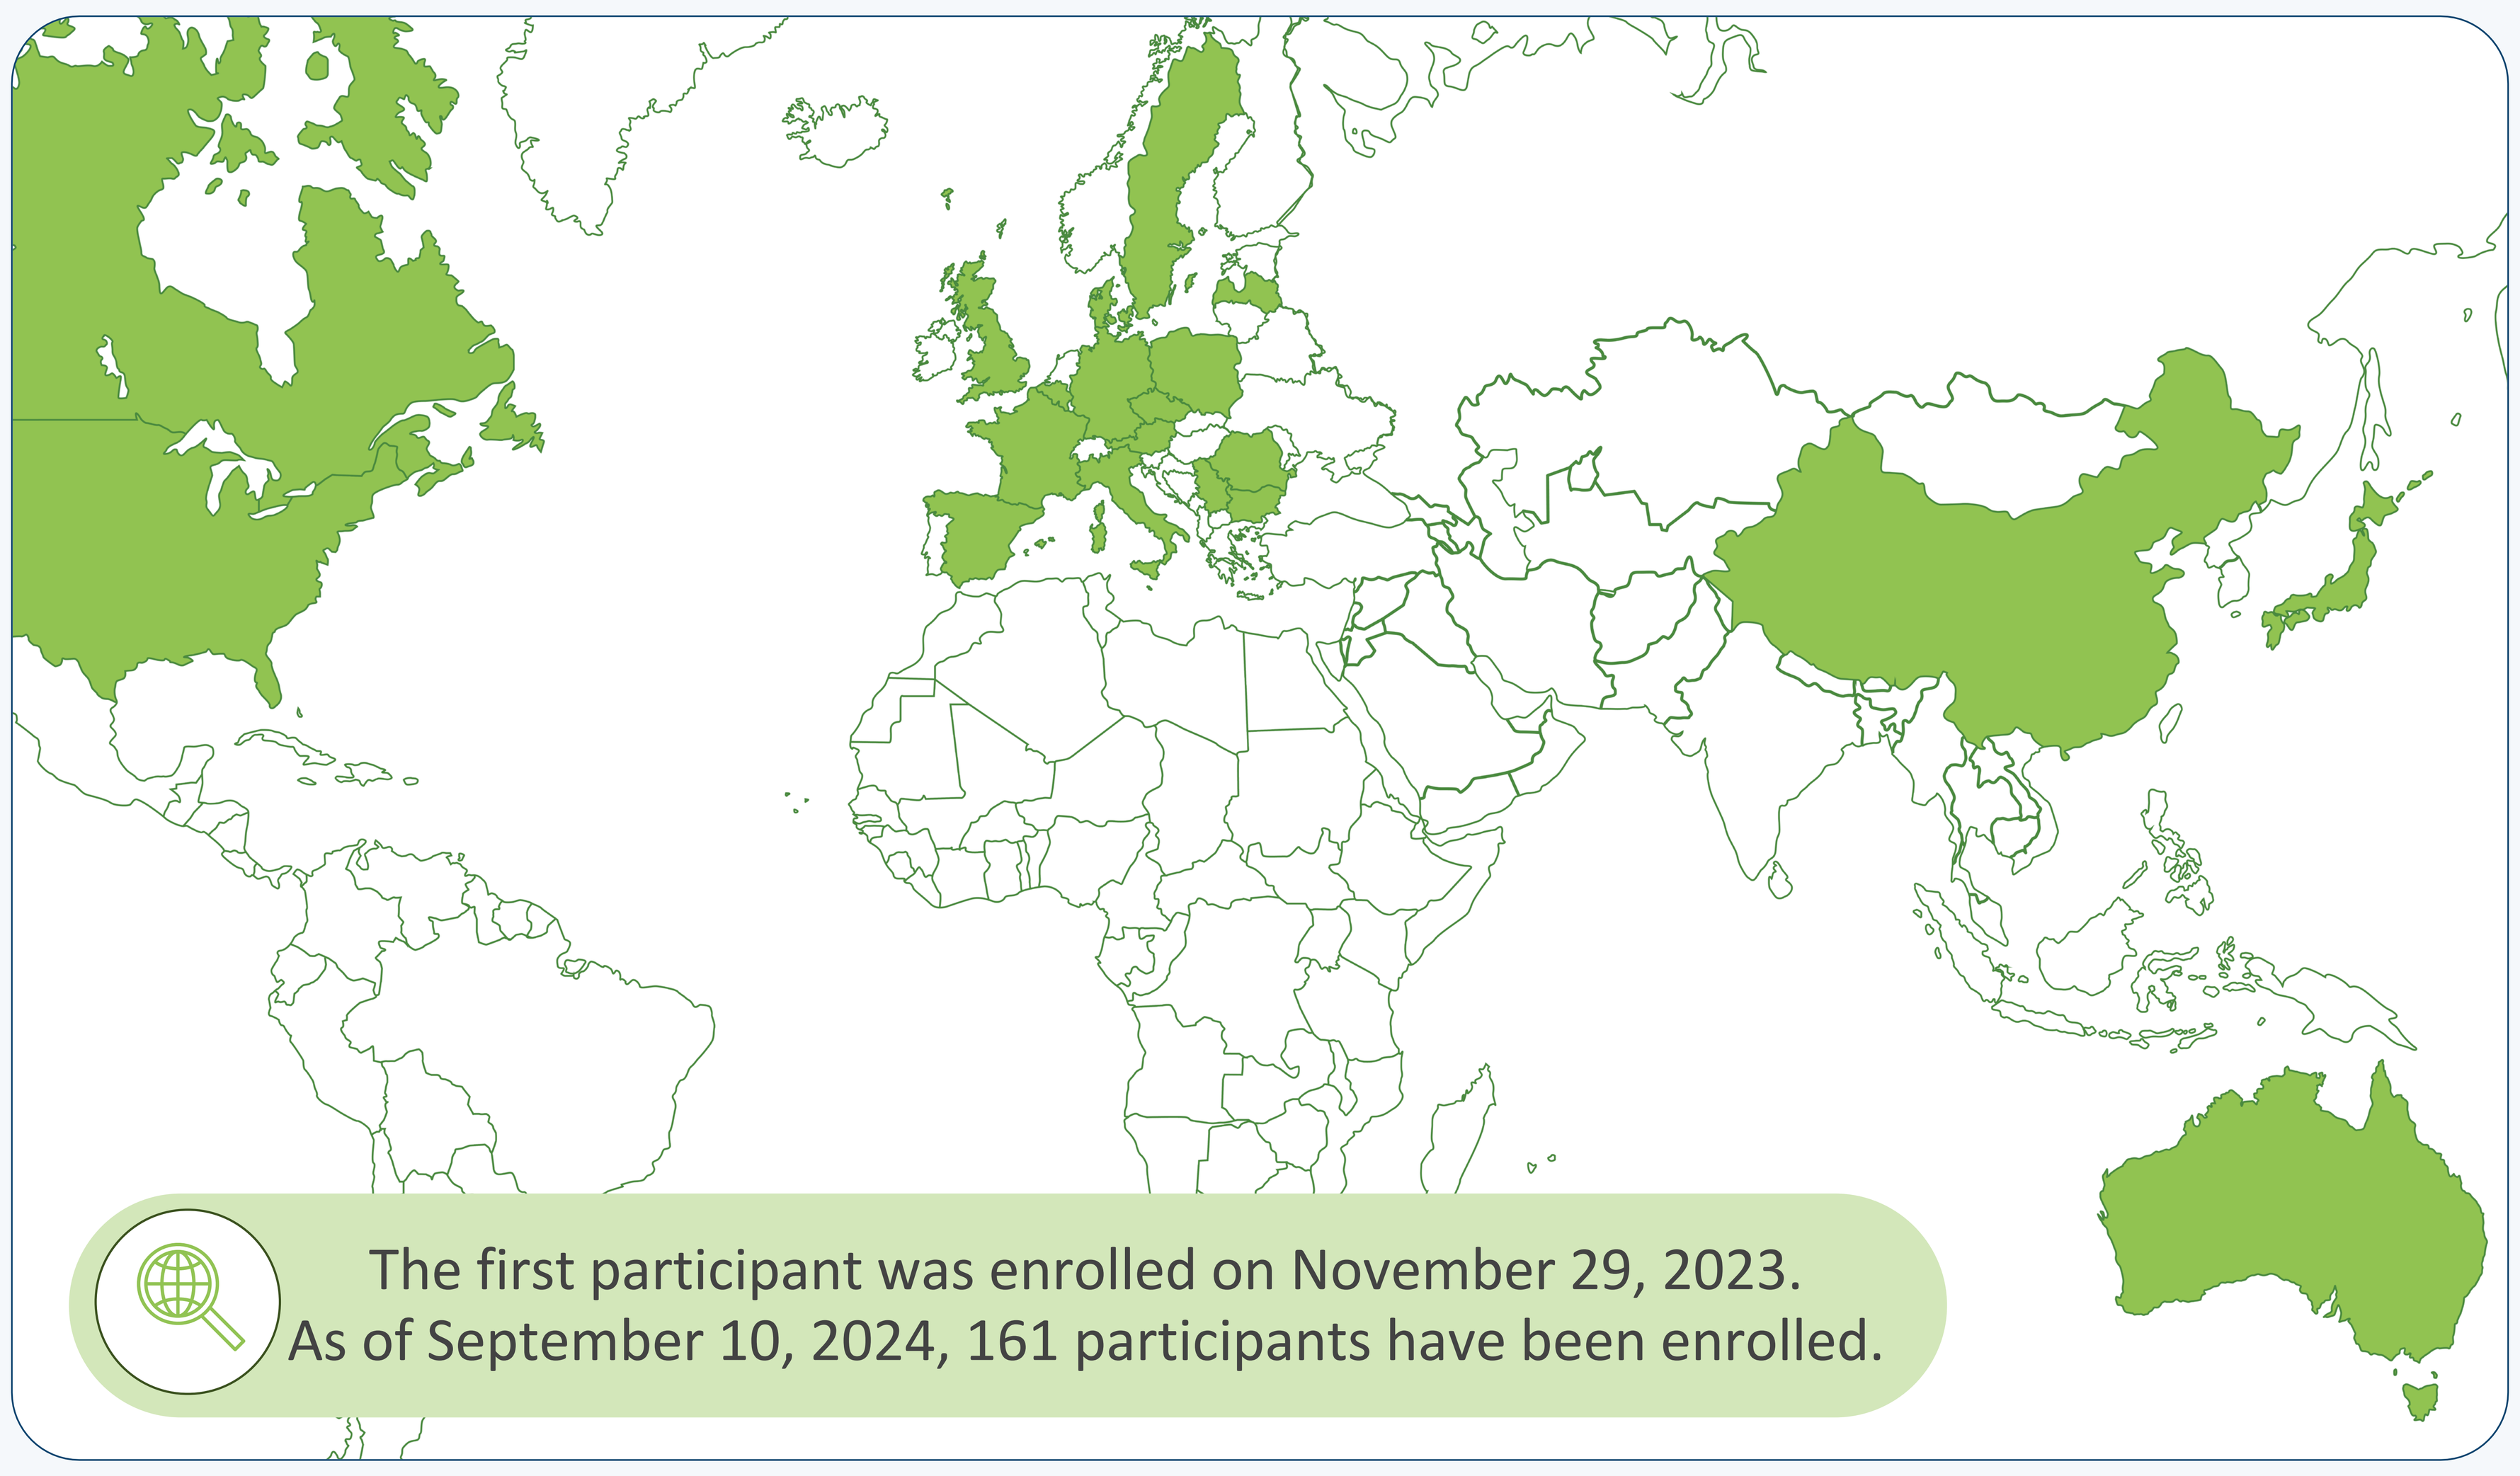

The first participant was enrolled on November 29, 2023.
As of September 10, 2024, 161 participants have been enrolled.

Australia
Austria
Belgium
Bulgaria
Canada
China
Czech Republic
Denmark
France
Germany
Italy
Japan
Latvia
Poland
Romania
Serbia
Spain
Sweden
United Kingdom
United States

STUDY OBJECTIVES AND ENDPOINTS

Assess the MMN disease course and management
Diagnosis parameters; Clinical characteristics; Initial symptoms; Limbs affected and number; Treatment parameters

Characterize MMN participant profiles
Demographics; Medical history; Prior and current therapy use

Estimate economic burden of disease for participants
Values and change over time in the HRPQ; Values over time for health care resource utilization

Assess outcome measures specific to MMN disease and their evolution over time
MMN-RODS, MMRC-10 sum score, Adjusted INCAT score

Estimate burden of MMN on participants' HRQoL
EQ-5D-5L; RT-FSS; CAP-PRI; PGI-S

Collect data on relevant disease biomarkers*
Serum levels of complement factors and complement activation; Serum titers of antiganglioside autoantibodies; NfL; Gene variants of complement regulatory proteins

*Optionally collected in specific countries where consent is provided by the participant.

KEY TAKEAWAYS

iMMersioN is an ongoing, global, prospective, longitudinal study aiming to provide a detailed view of the impact of MMN and its treatment on participants in a real-world setting

The study will examine clinical outcomes, disease course, HRQoL, and resource utilization over 24 months in ~200 adult participants with MMN

Presented at the 2024 Neuromuscular Study Group (NMSG) Annual Scientific Meeting; September 20–22, 2024; Tarrytown, NY, USA

ABBREVIATIONS

CAP-PRI, Chronic Acquired Polyneuropathy Patient-Reported Index; EQ-5D-5L, EuroQoL 5-Dimension 5-Level; HRPQ, Health-Related Productivity Questionnaire; HRQoL, Health-Related Quality of Life; IgM, immunoglobulin M; INCAT, Inflammatory Neuropathy Cause and Treatment; MMN, multifocal motor neuropathy; MMN-RODS, Rasch-built Overall Disability Scale for MMN; MMRC, Modified Medical Research Council; NfL, Neurofilament light chain; PGI-S, Patient Global Impression of Severity; RT-FSS, Rasch-Transformed Fatigue Severity Scale.

DISCLOSURES AND ACKNOWLEDGMENTS

SP: lecture honoraria from ADOC, argenx, Berlin-Chemie Menarini, Mylan, Pfizer, Remedica, Salveo, Teva Actavis, Viatris, and Wörwag; research grants from argenx, Kedrion Biopharma and Octapharma; consultant fees from argenx, Dianthus Therapeutics and Mylan; and travel grants from ADOC, Berlin-Chemie Menarini, Kedrion Biopharma, Medis, Octapharma, Pfizer, Roche, Sanofi Genzyme, Teva Actavis and Wörwag; and reports no other conflicts of interest outside or related to this work; **LQ**: research grants from CIBERER, Fundació La Marató, GBS–CIDP Foundation International, Grifols, Instituto de Salud Carlos III – Ministry of Economy and Innovation (Spain) and UCB; speaker or expert testimony honoraria from Alnylam, Annexon, argenx, Avilar Therapeutics, Biogen, CSL Behring, Janssen, LFB, Lundbeck, Merck, Novartis, Octapharma, Roche, Sanofi Genzyme, and UCB; serves at Clinical Trial Steering Committee for Sanofi Genzyme; and was Principal Investigator for UCB's CIDP01 trial; **SA**: nothing to disclose; **JW**: argenx, Takeda; **IVW**, **EP**, **IVH**, **MH**, **CA-B**: employees of argenx; **GS**, **MV**: consultants for argenx and PPD; **OVS**: consultant for argenx; **JAA**: consulting honoraria from Akcea, Alexion, Alnylam, Annexon, argenx, CSL Behring, Grifols, Immunovant, ImmuPharma, Johnson & Johnson, Pfizer and Takeda.

This study is sponsored by argenx. Medical writing support was provided by Envision Pharma Group, funded by argenx.

REFERENCES

1. Yeh WZ, et al. *J Neurol Neurosurg Psychiatry*. 2020;91:140–8. **2.** Budding K, et al. *Neurol Neuroimmunol Neuroinflamm*. 2021;9:e1107. **3.** Harschnitz O, et al. *J Clin Immunol*. 2014;34(suppl 1):S112–9. **4.** Katz J, et al. First global multifocal motor neuropathy (MMN) quality of life (QoL) patient survey identifies needs in education and treatment. Accessed September 10, 2024. http://www.neuropathyaction.org/downloads/MMN_article%209-26-2016.pdf/.

SCAN ME
to access the full poster